First-in-Human Phase 1 Study of a Novel Claudin 6 (CLDN6) Targeted Antibody Drug Conjugate (ADC) TORL-1-23

756P

G.E. Konecny[1], A. Wahner Hendrickson[2], B. Winterhoff[3], A. Machado[4], C. Chander[5], T. Hawkins[3], S. Davenport[6], S. Bilic[7], L.L. Miller[8], A. Chung[8], M. Press[9], S. Letrent[10], D. Slamon[11]

[1]Department of Medicine, UCLA Westwood Oncology Hematology, Los Angeles, CA, [2]Medical Oncology, Mayo Clinic, Rochester, MN, [3]Division of Gynecologic Oncology, University of Minnesota, Minneapolis, MN, [4]Medical, TRIO – Translational Research in Oncology, Montevideo, Uruguay, [5]Department of Medicine, Jonsson Comprehensive Cancer Center, UCLA, Los Angeles, CA, [6]Department of Medicine, USC Norris Comprehensive Cancer Center, Los Angeles, CA, [7]Clinical Pharmacology, Vanadro, L.L.C., Waukee, IA, [8]Clinical Operations, TRIO-US, Los Angeles, CA, [9]Department of Pathology, USC Keck School of Medicine, Los Angeles, CA, [10]Research and Development, TORL BioTherapeutics, Culver City, CA, [11]Division of Hematology-Oncology, UCLA David Geffen School of Medicine, Los Angeles, CA

Introduction

- CLDN6 is a transmembrane protein of tight junctions important for cell-to-cell connectivity
 - Implicated in the initiation, progression, and metastasis of some cancers
 - Expressed at high levels in multiple cancers with little to no expression in normal tissues

Figure 1. CLDN6 Expression in Cancer

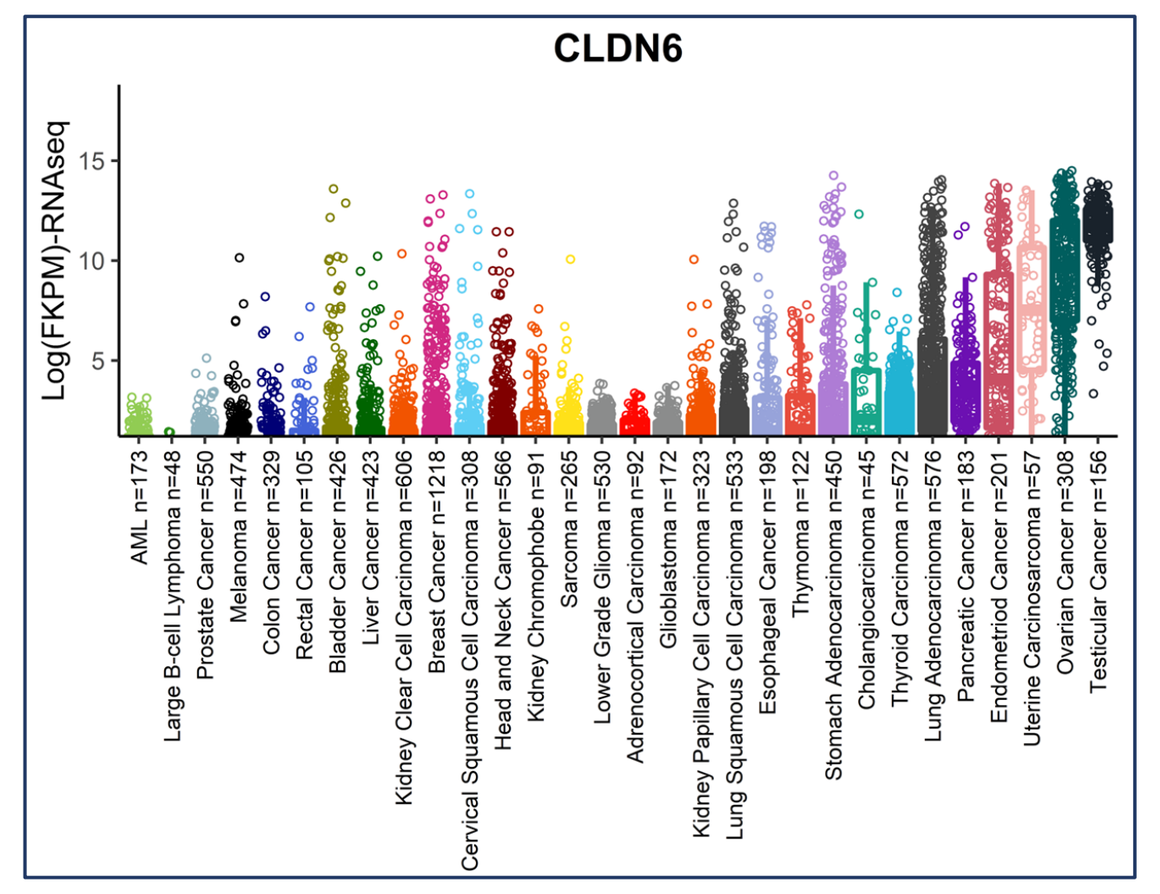
CLDN6
Log(FKPM)-RNAseq

Figure 2. CLDN6 Expression in Normal Tissue

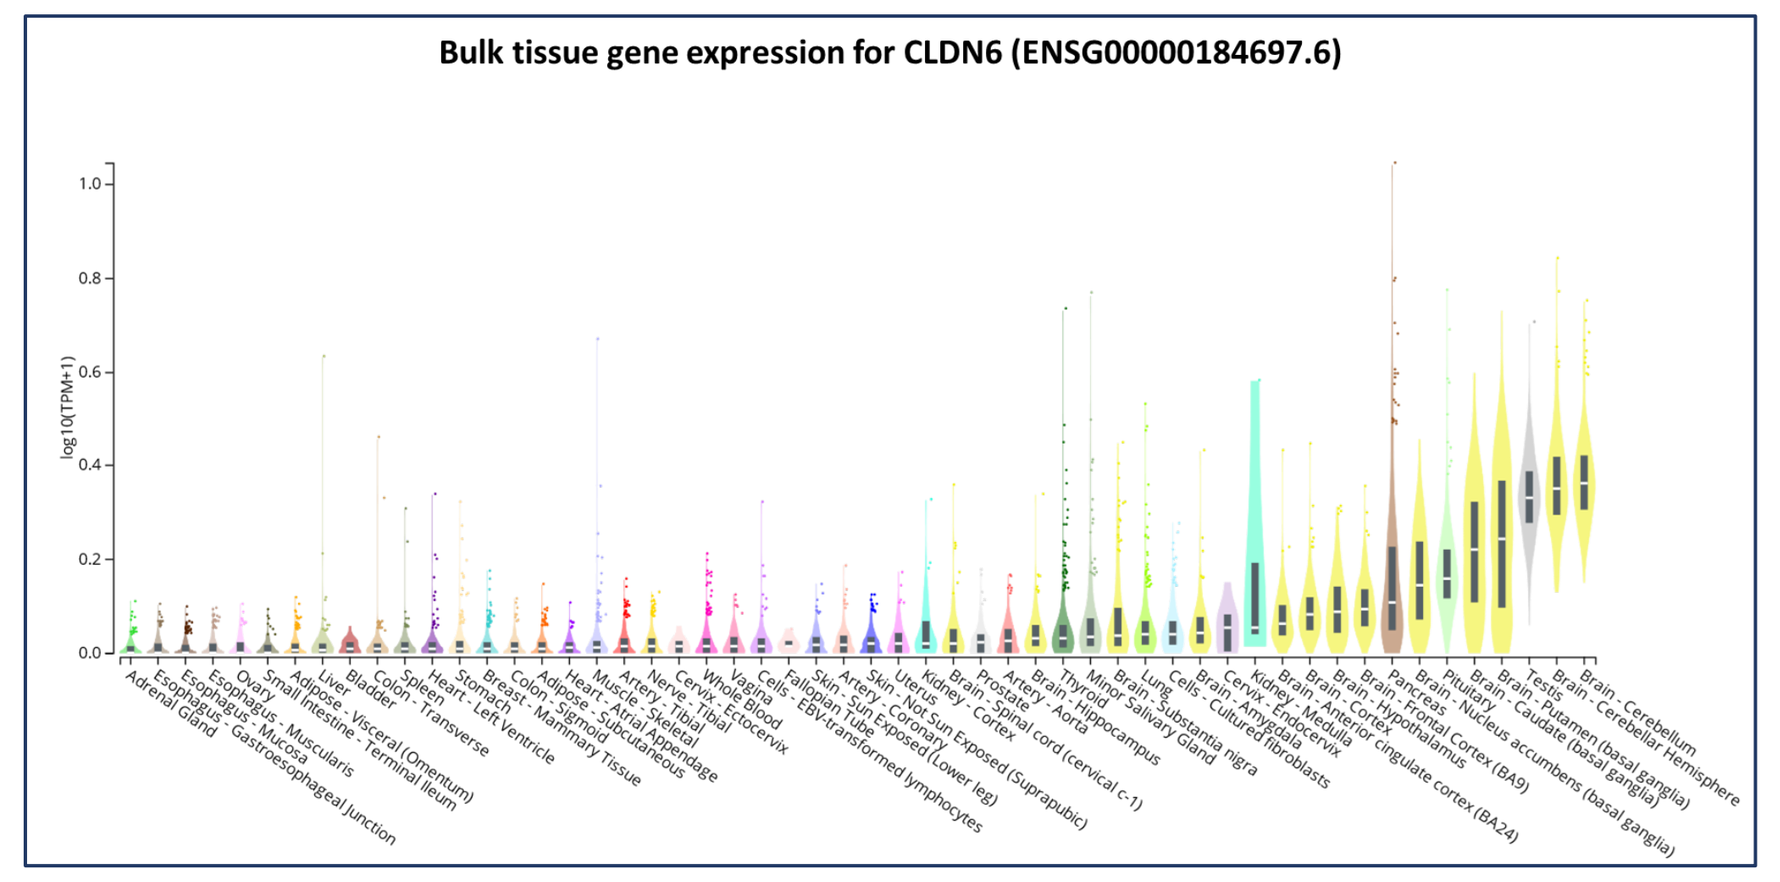
Bulk tissue gene expression for CLDN6 (ENSG00000184697.6)

Figure 3. CLDN6 is a Unique Subset of Ovarian Cancer

Total Ovarian Patient Samples (n=308) **CLDN6hi Patient Samples (n=77)**

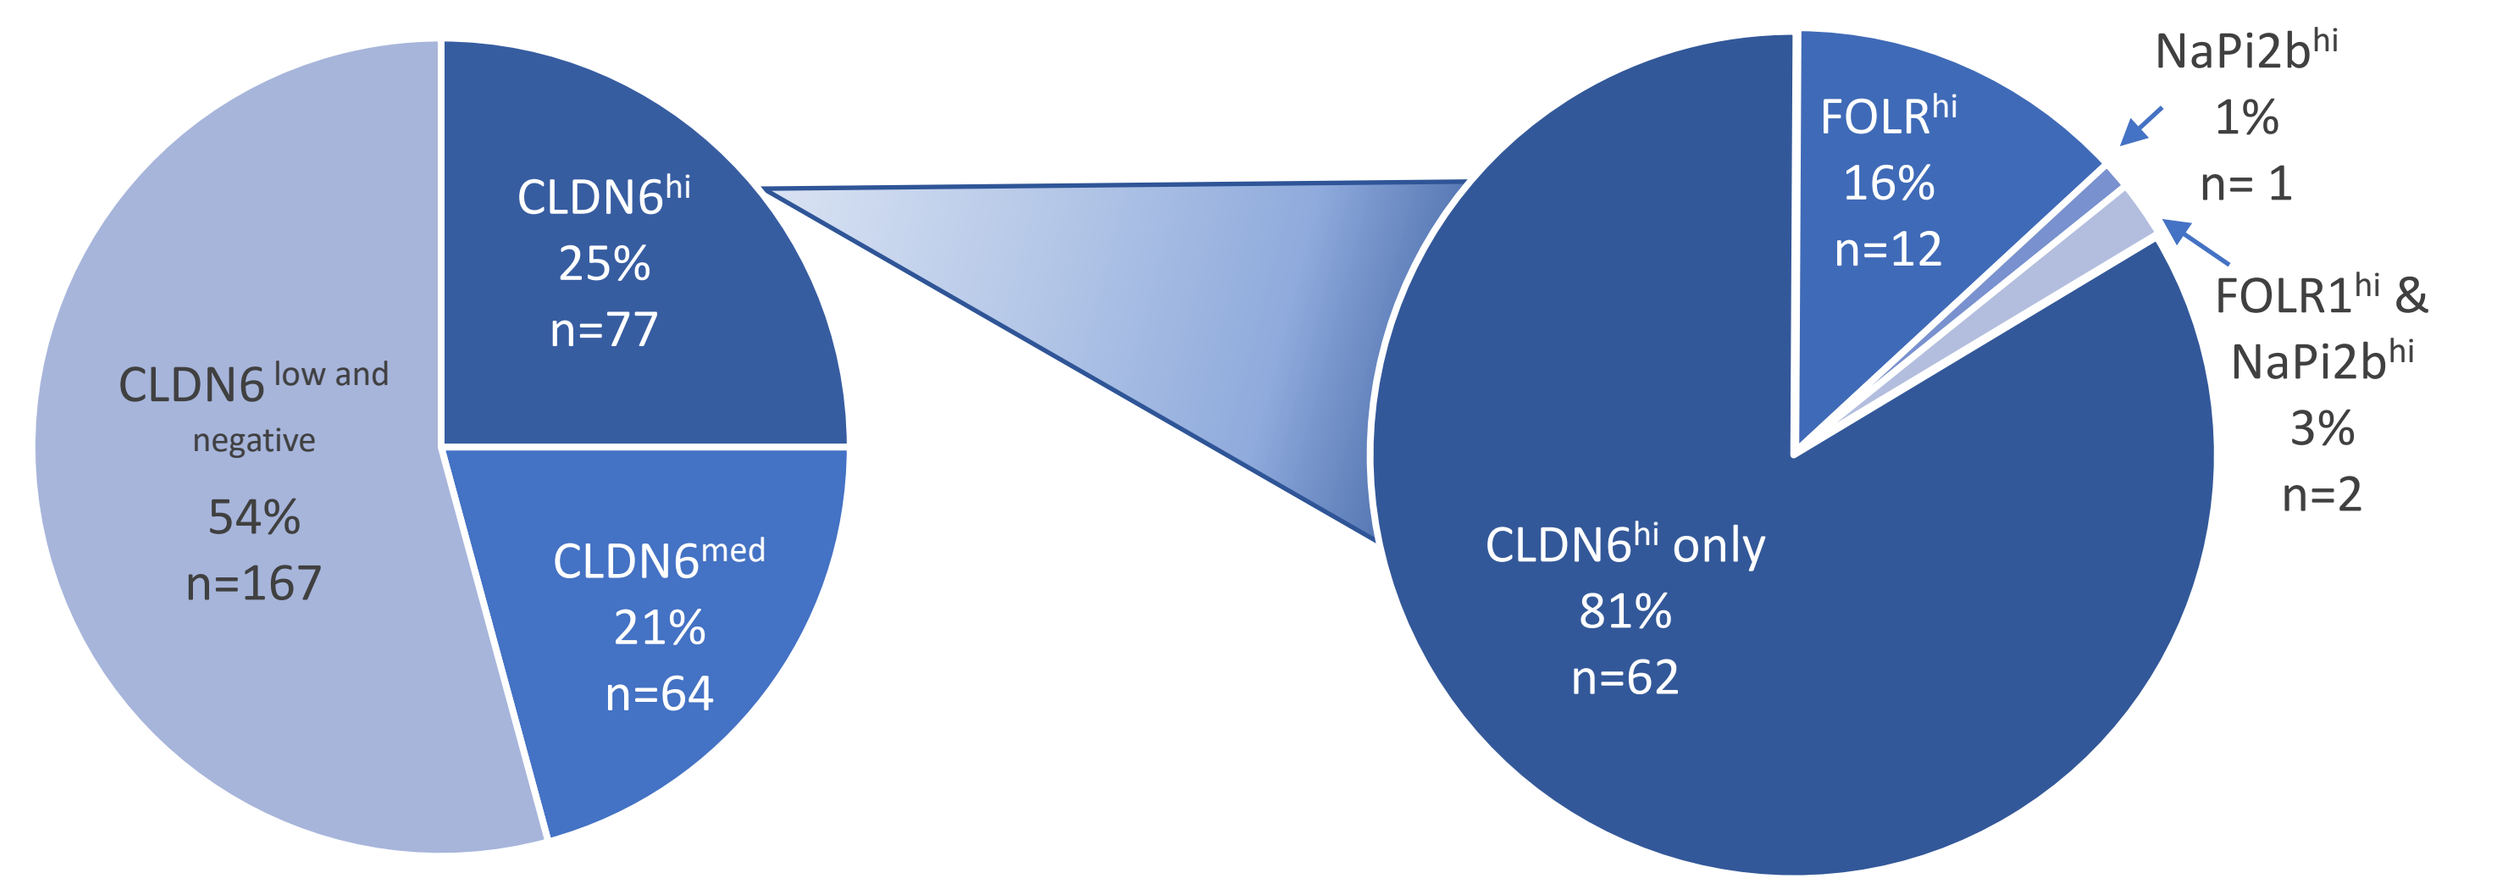
CLDN6hi
25%
n=77
CLDN6 low and negative
54%
n=167
CLDN6med
21%
n=64
FOLRhi
16%
n=12
NaPi2bhi
1%
n= 1
FOLR1hi & NaPi2bhi
3%
n=2
CLDN6hi only
81%
n=62

- The top 25% of CLDN6 expression (RNA based) was used as the cut off for high expression and within that population the number of patients that reached the 25% expression threshold for FOLR1a and NaPi2b was assessed and counted.

Pharmacology of TORL-1-23

- TORL-1-23 ADC is a fully humanized IgG1 (TORL-1-23-MAb) linked to MMAE through a cathepsin hydrolysable dipeptide VC linker (vc-MMAE)
- TORL-1-23 binds to CLDN6 with a high affinity and specificity with rapid ADC-CLND6 complex internalization and release of an MMAE payload which disrupts the microtubule network leading to cell cycle arrest and apoptosis. Fc effector function (eg, ADCC) contributes only ~10% to the mechanism of TORL-1-23.
- TORL-1-23 is in development for the treatment of CLDN6-positive cancers including ovarian and non-small cell lung cancer (NSCLC)

Figure 4. Specific Binding of TORL-1-23 to CLDN Family Members

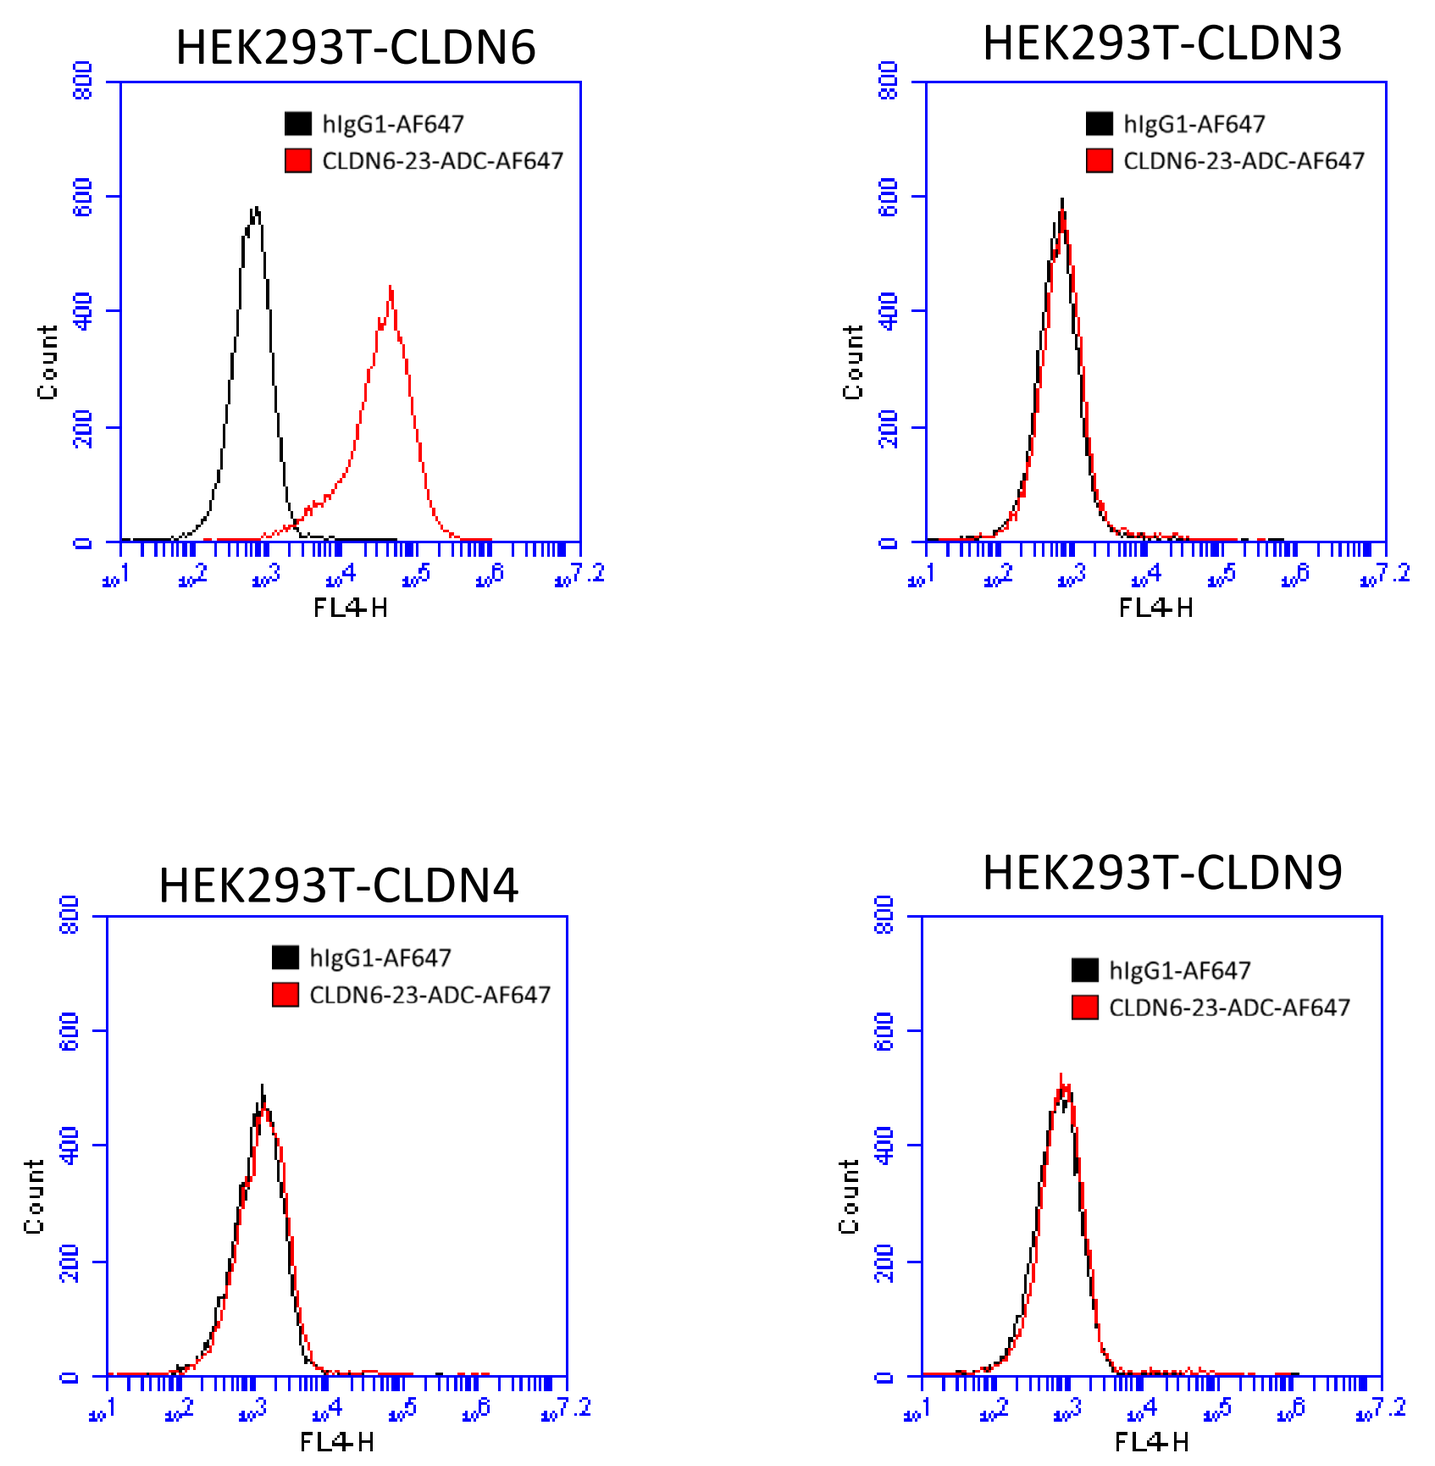
HEK293T-CLDN6
HEK293T-CLDN3
HEK293T-CLDN4
HEK293T-CLDN9
hIgG1-AF647
CLDN6-23-ADC-AF647
Count
FL4-H

- Binding of TORL-1-23 (5µg/ml) in HEK293T cells artificially overexpressing CLDN3, CLDN4, CLDN6 or CLDN9 by flow cytometry.
- TORL-1-23 exhibited robust, selective binding to only CLDN6-overexpressing cells

Methods

TORL123-001 (TRIO-049) Study Objectives

- Characterize the safety, tolerability, and DLT and determine the MTD and RP2D for TORL-1-23
- Characterize the PK of TORL-1-23 and its breakdown products (MMAE and TORL-1-23 MAB)
- Assess the preliminary antitumor activity of TORL-1-23 in participants with advanced cancer
- Assess the immunogenicity of TORL-1-23

Figure 5. TORL123-001 (TRIO-049) Phase 1 Study Design

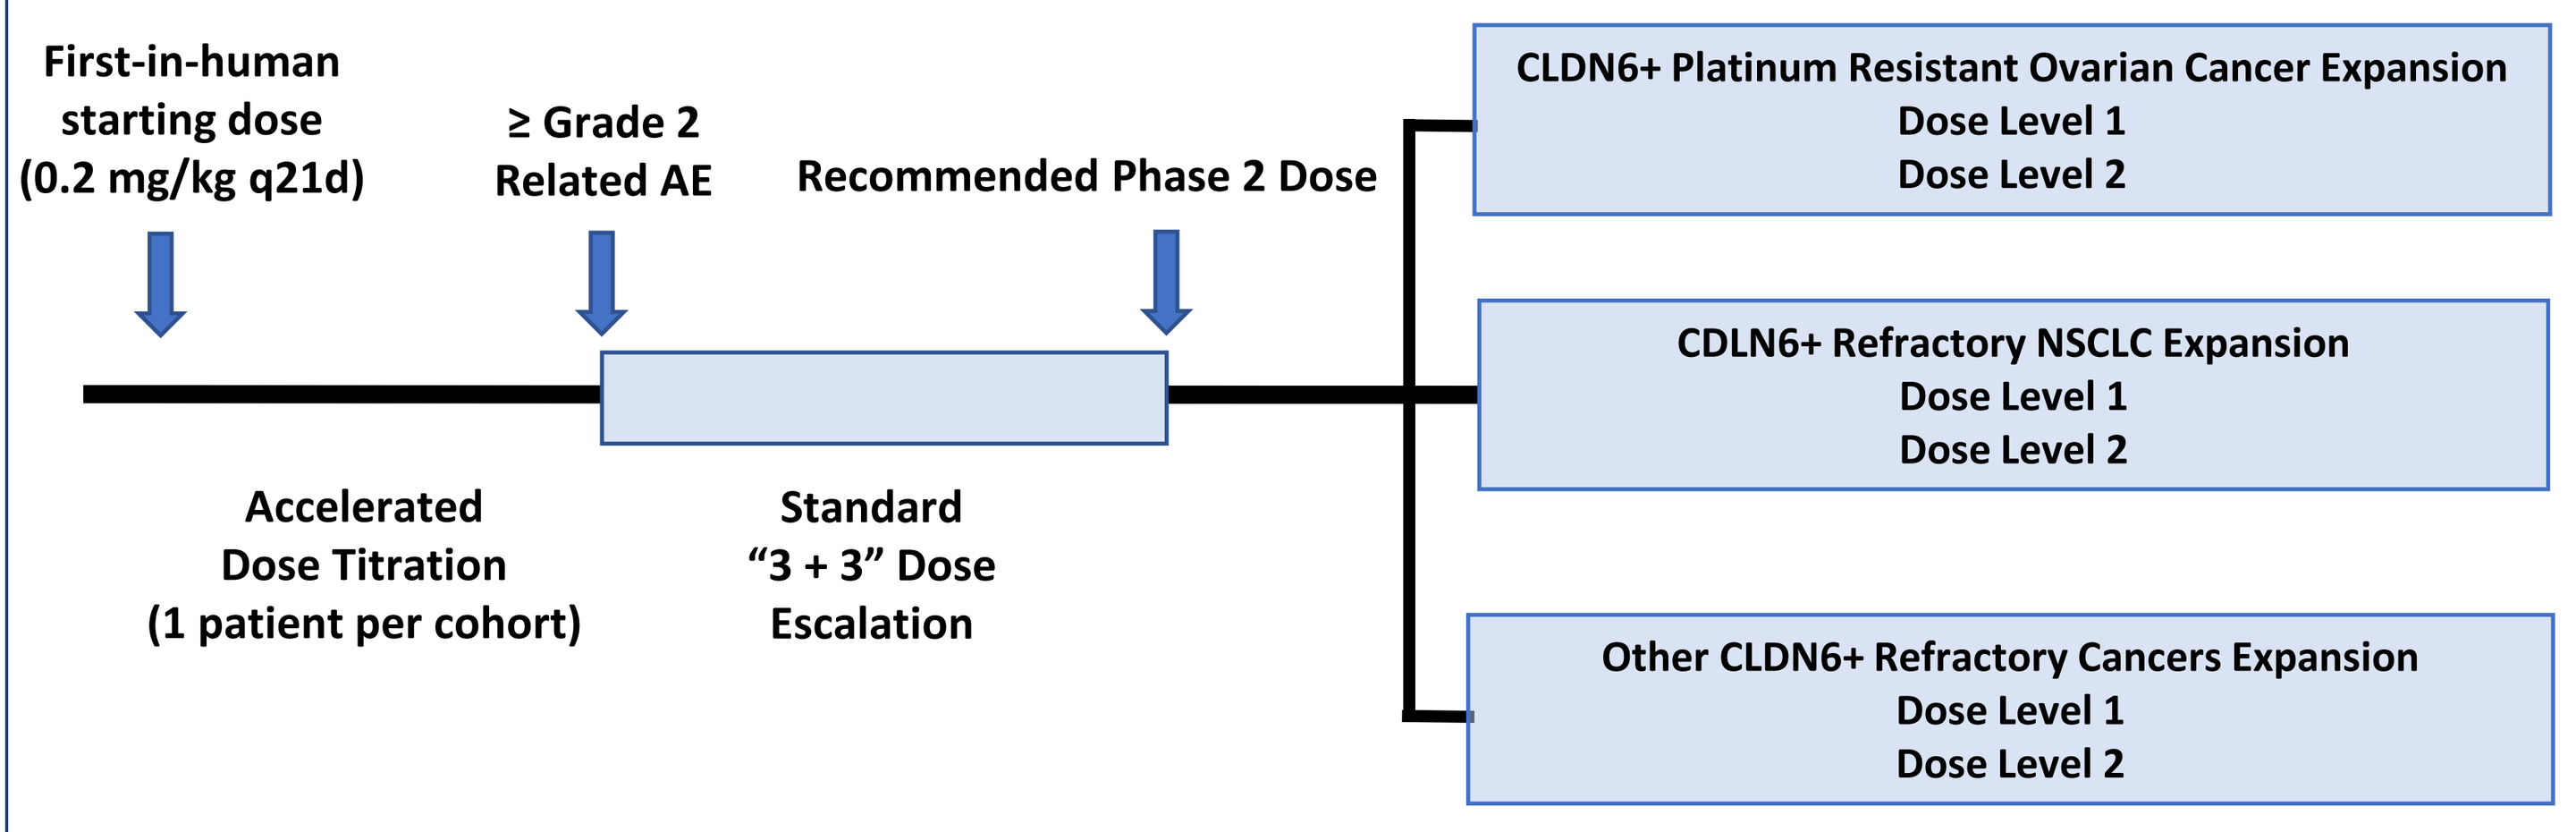
First-in-human starting dose (0.2 mg/kg q21d)
≥ Grade 2 Related AE
Recommended Phase 2 Dose
Accelerated Dose Titration (1 patient per cohort)
Standard "3 + 3" Dose Escalation
CLDN6+ Platinum Resistant Ovarian Cancer Expansion
Dose Level 1
Dose Level 2
CDLN6+ Refractory NSCLC Expansion
Dose Level 1
Dose Level 2
Other CLDN6+ Refractory Cancers Expansion
Dose Level 1
Dose Level 2

Dose Cohort	1	2	3	4	5	6	7	8	9	10
Dose (mg/kg)	0.2	0.4	0.8	1.0	1.3	1.7	2.0	2.4	3.0	3.0+GCSF

NCT05103683
Study Sponsor: TORL Biotherapeutics

Study Design

- Accelerated dose finding design in refractory tumors with expansion cohorts in CLDN6+ cancers
- TORL-1-23 given IV Q3W until disease progression, unacceptable toxicity, or study withdrawal
- Intra-patient dose escalation allowed

Key Study Eligibility

- Dose Finding: Advanced solid tumor unresponsive to standard therapies
- Dose Expansion: CLDN6+ cancers as determined by IHC
 - Platinum-resistant ovarian cancer, refractory NSCLC, other CLDN6+ refractory cancers
- Measurable disease, adequate organ function, ECOG performance status 0-1
- <Grade 2 toxicity from prior therapies at baseline

Dose Limiting Toxicity (DLT) Criteria

- Any death not clearly due to underlying disease or extraneous causes
- ≥Grade 4 hematologic toxicity, ≥Grade 3 febrile neutropenia, ≥Grade 3 thrombocytopenia with bleeding, ≥Grade 3 non-hematologic toxicity with exceptions for gastrointestinal events, changes in electrolytes, liver enzyme changes <14 days, or dose delays >14 days

Table 1. Demographics

Dose, mg/kg	0.2	0.4	0.8	1.0	1.3	1.7	2.0	2.4	3.0	3+GCSF	Total
N	1	1	1	3	3	5[a]	5[a]	6[a]	11[a]	6[a]	42
Age, years (range)	56 -	44 -	57 -	50 (30-69)	55 (48-70)	62 (52-66)	51 (27-65)	66 (59-74)	59 (26-78)	64 (53-74)	57 (26-78)
Gender, male/female	0/1	1/0	0/1	1/2	0/3	0/5	1/4	0/6	2/9	0/6	5/37
Cancer type, n											
Ovarian	1	0	1	2	3	4	4	4	8	3	30
Testicular	0	1	0	1	0	0	1	0	2	0	5
Endometrial	0	0	0	0	0	1	0	2	1	3	7
Median number of prior treatments (range)	6 -	3 -	3 -	5 (1-8)	4 (3-6)	5 (3-7)	6 (3-9)	4 (1-6)	3 (2-4)	3 (2-3)	4 (1-9)
CLDN6 Positive by IHC n/N	1/1	1/1	1/1	3/3	3/3	3/5	5/5	6/6	11/11	6/6	40/42

[a]Cohort expanded to further characterize and guide dose selection. Datacut 29SEP2023

Results

Table 2. Treatment Related Adverse Events Occurring in 10% of Patients or Greater

Preferred Term	Dose (mg/kg)	0.2 N=1	0.4 N=1	0.8 N=1	1.0 N=3	1.3 N=3	1.7 N=5	2.0 N=5	2.4 N=6	3.0 N=11	3.0 +GCSF N=6	Total N=42
Patients with any TRAE, n (%)		1 (100)	1 (100)	1 (100)	2 (66)	1 (33)	3 (60)	3 (60)	6 (100)	11 (100)	2 (40)	31 (74)
Alopecia	All Grades	1	-	-	-	-	-	1	4	7	1	14
	G1	1	-	-	-	-	-	1	2	2	1	7
	G2	-	-	-	-	-	-	-	2	5	-	7
Anemia	All Grades	-	-	-	1	-	1	-	3	8	1	14
	G1	-	-	-	-	-	1	-	2	4	1	8
	G2	-	-	-	-	-	-	-	-	3	-	3
	G3	-	-	-	1	-	-	-	1	1	-	3
Arthralgia	All Grades	-	-	-	-	-	-	-	-	6	-	6
	G1	-	-	-	-	-	-	-	-	3	-	3
	G2	-	-	-	-	-	-	-	-	2	-	2
	G3	-	-	-	-	-	-	-	-	1	-	1
Aspartate aminotransferase increased	All Grades	-	-	-	-	-	-	-	1	3	1	5
	G1	-	-	-	-	-	-	-	1	3	1	5
Constipation	All Grades	-	-	-	-	-	1	-	-	4	-	5
	G1	-	-	-	-	-	1	-	-	2	-	3
	G2	-	-	-	-	-	-	-	-	2	-	2
Decreased appetite	All Grades	-	-	-	-	-	1	-	2	2	1	6
	G1	-	-	-	-	-	-	-	2	1	1	4
	G2	-	-	-	-	-	1	-	-	1	-	2
Fatigue	All Grades	-	-	1	1	-	3	-	1	8	2	16
	G1	-	-	1	1	-	1	-	-	5	2	10
	G2	-	-	-	-	-	2	-	1	3	-	6
Nausea	All Grades	-	-	-	-	-	2	-	1	5	2	10
	G1	-	-	-	-	-	1	-	1	3	1	6
	G2	-	-	-	-	-	1	-	-	2	1	4
Neuropathy	All Grades	-	1	1	1	-	1	1	2	6	-	13
	G1	-	1	1	1	-	1	-	1	3	-	8
	G2	-	-	-	-	-	-	1	1	2	-	4
	G3	-	-	-	-	-	-	-	-	1	-	1
Neutropenia	All Grades	-	-	-	-	-	-	2	2	5	-	9
	G2	-	-	-	-	-	-	1	-	-	-	1
	G3	-	-	-	-	-	-	1	1	1	-	3
	G4	-	-	-	-	-	-	-	1	4	-	5
Pneumonia	All Grades	-	-	-	-	-	-	-	1	-	-	1
	G5	-	-	-	-	-	-	-	1	-	-	1
White blood cell count decreased	All Grades	-	-	-	-	-	-	-	2	7	-	9
	G1	-	-	-	-	-	-	-	2	2	-	4
	G2	-	-	-	-	-	-	-	-	4	-	4
	G4	-	-	-	-	-	-	-	-	1	-	1
Dose Limiting Toxicities (DLT)	All	0	0	0	0	0	0	0	0	3	0	3

Datacut 29SEP2023

Figure 6. Average TORL-1-23 Concentrations by Dose: ***ADC Conjugate, Total Mab, and MMAE Overlay for 0.2 to 2.4 mg/kg (3.0 mg/kg pending)***

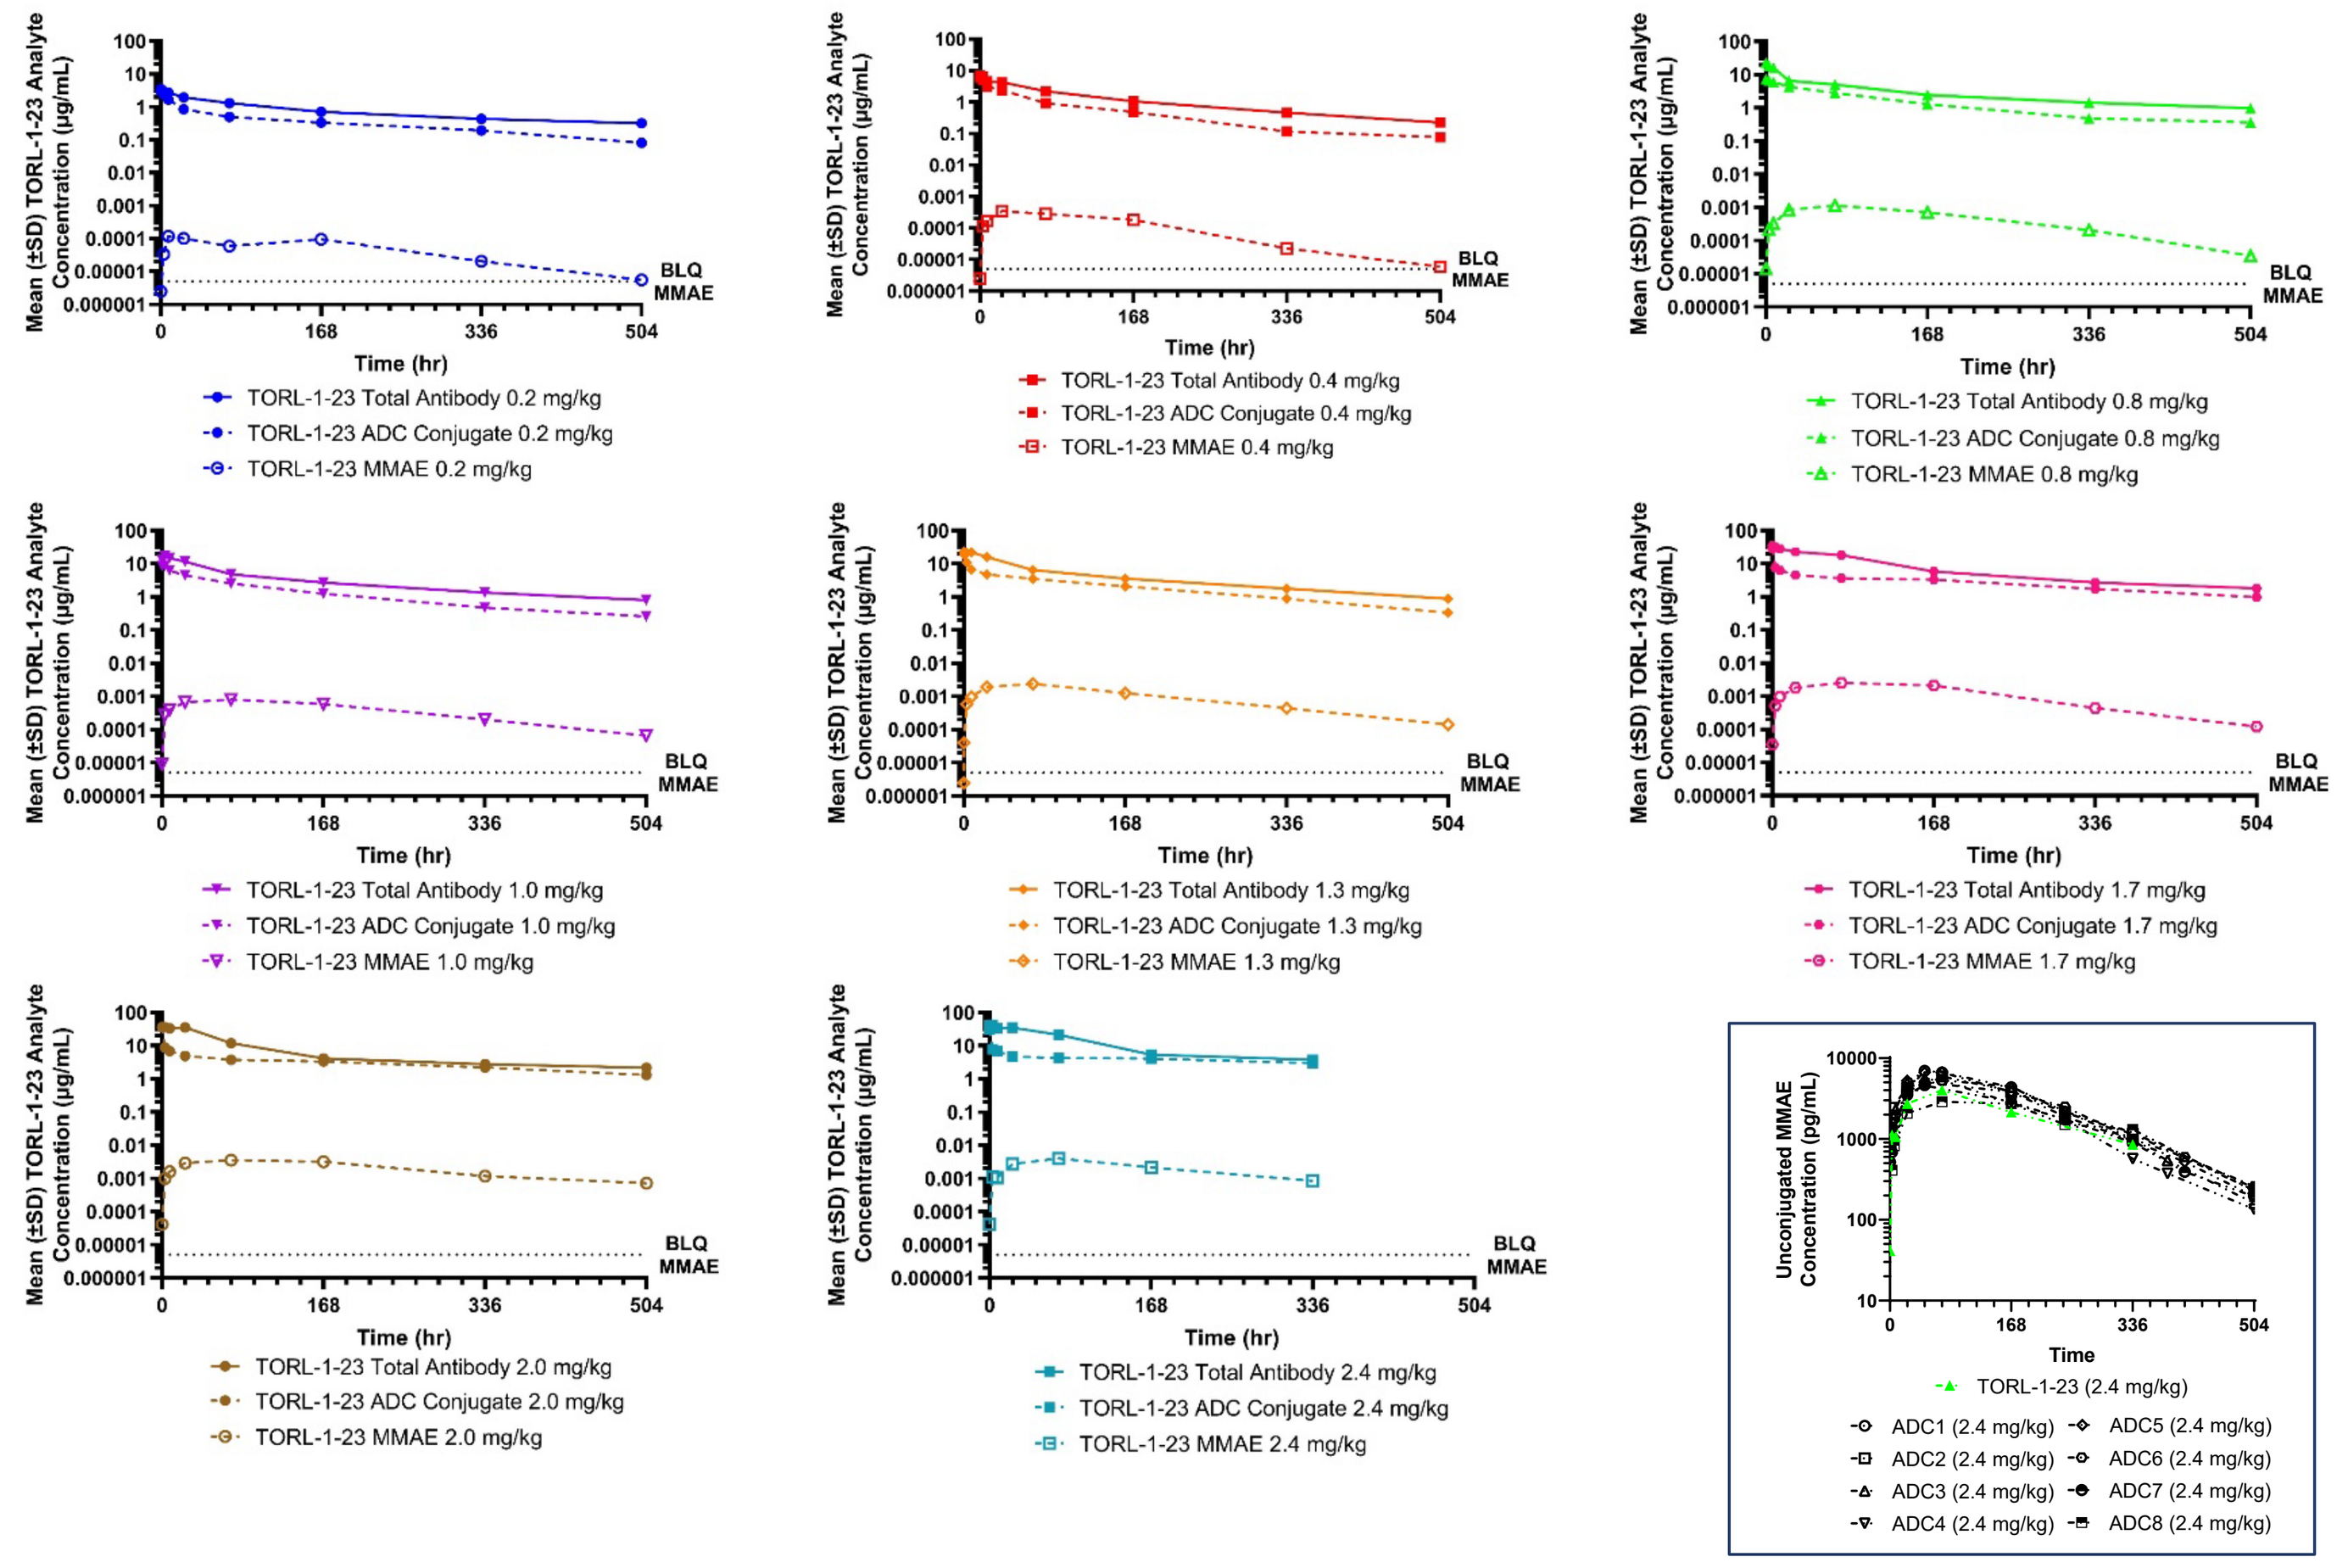
Mean (±SD) TORL-1-23 Analyte Concentration (µg/mL)
Time (hr)
BLQ MMAE
TORL-1-23 Total Antibody 0.2 mg/kg
TORL-1-23 ADC Conjugate 0.2 mg/kg
TORL-1-23 MMAE 0.2 mg/kg
TORL-1-23 Total Antibody 0.4 mg/kg
TORL-1-23 ADC Conjugate 0.4 mg/kg
TORL-1-23 MMAE 0.4 mg/kg
TORL-1-23 Total Antibody 0.8 mg/kg
TORL-1-23 ADC Conjugate 0.8 mg/kg
TORL-1-23 MMAE 0.8 mg/kg
TORL-1-23 Total Antibody 1.0 mg/kg
TORL-1-23 ADC Conjugate 1.0 mg/kg
TORL-1-23 MMAE 1.0 mg/kg
TORL-1-23 Total Antibody 1.3 mg/kg
TORL-1-23 ADC Conjugate 1.3 mg/kg
TORL-1-23 MMAE 1.3 mg/kg
TORL-1-23 Total Antibody 1.7 mg/kg
TORL-1-23 ADC Conjugate 1.7 mg/kg
TORL-1-23 MMAE 1.7 mg/kg
TORL-1-23 Total Antibody 2.0 mg/kg
TORL-1-23 ADC Conjugate 2.0 mg/kg
TORL-1-23 MMAE 2.0 mg/kg
TORL-1-23 Total Antibody 2.4 mg/kg
TORL-1-23 ADC Conjugate 2.4 mg/kg
TORL-1-23 MMAE 2.4 mg/kg
Unconjugated MMAE Concentration (pg/mL)
Time
TORL-1-23 (2.4 mg/kg)
ADC1 (2.4 mg/kg)
ADC2 (2.4 mg/kg)
ADC3 (2.4 mg/kg)
ADC4 (2.4 mg/kg)
ADC5 (2.4 mg/kg)
ADC6 (2.4 mg/kg)
ADC7 (2.4 mg/kg)
ADC8 (2.4 mg/kg)

- Human PK characteristics of TORL-1-23 were compared to results from 8 other vc-MMAE ADCs (Li et al. *MAbs* 2020).

Figure 7. Tumor Response by RECIST 1.1 Dose Levels 0.2 to 3 mg/kg

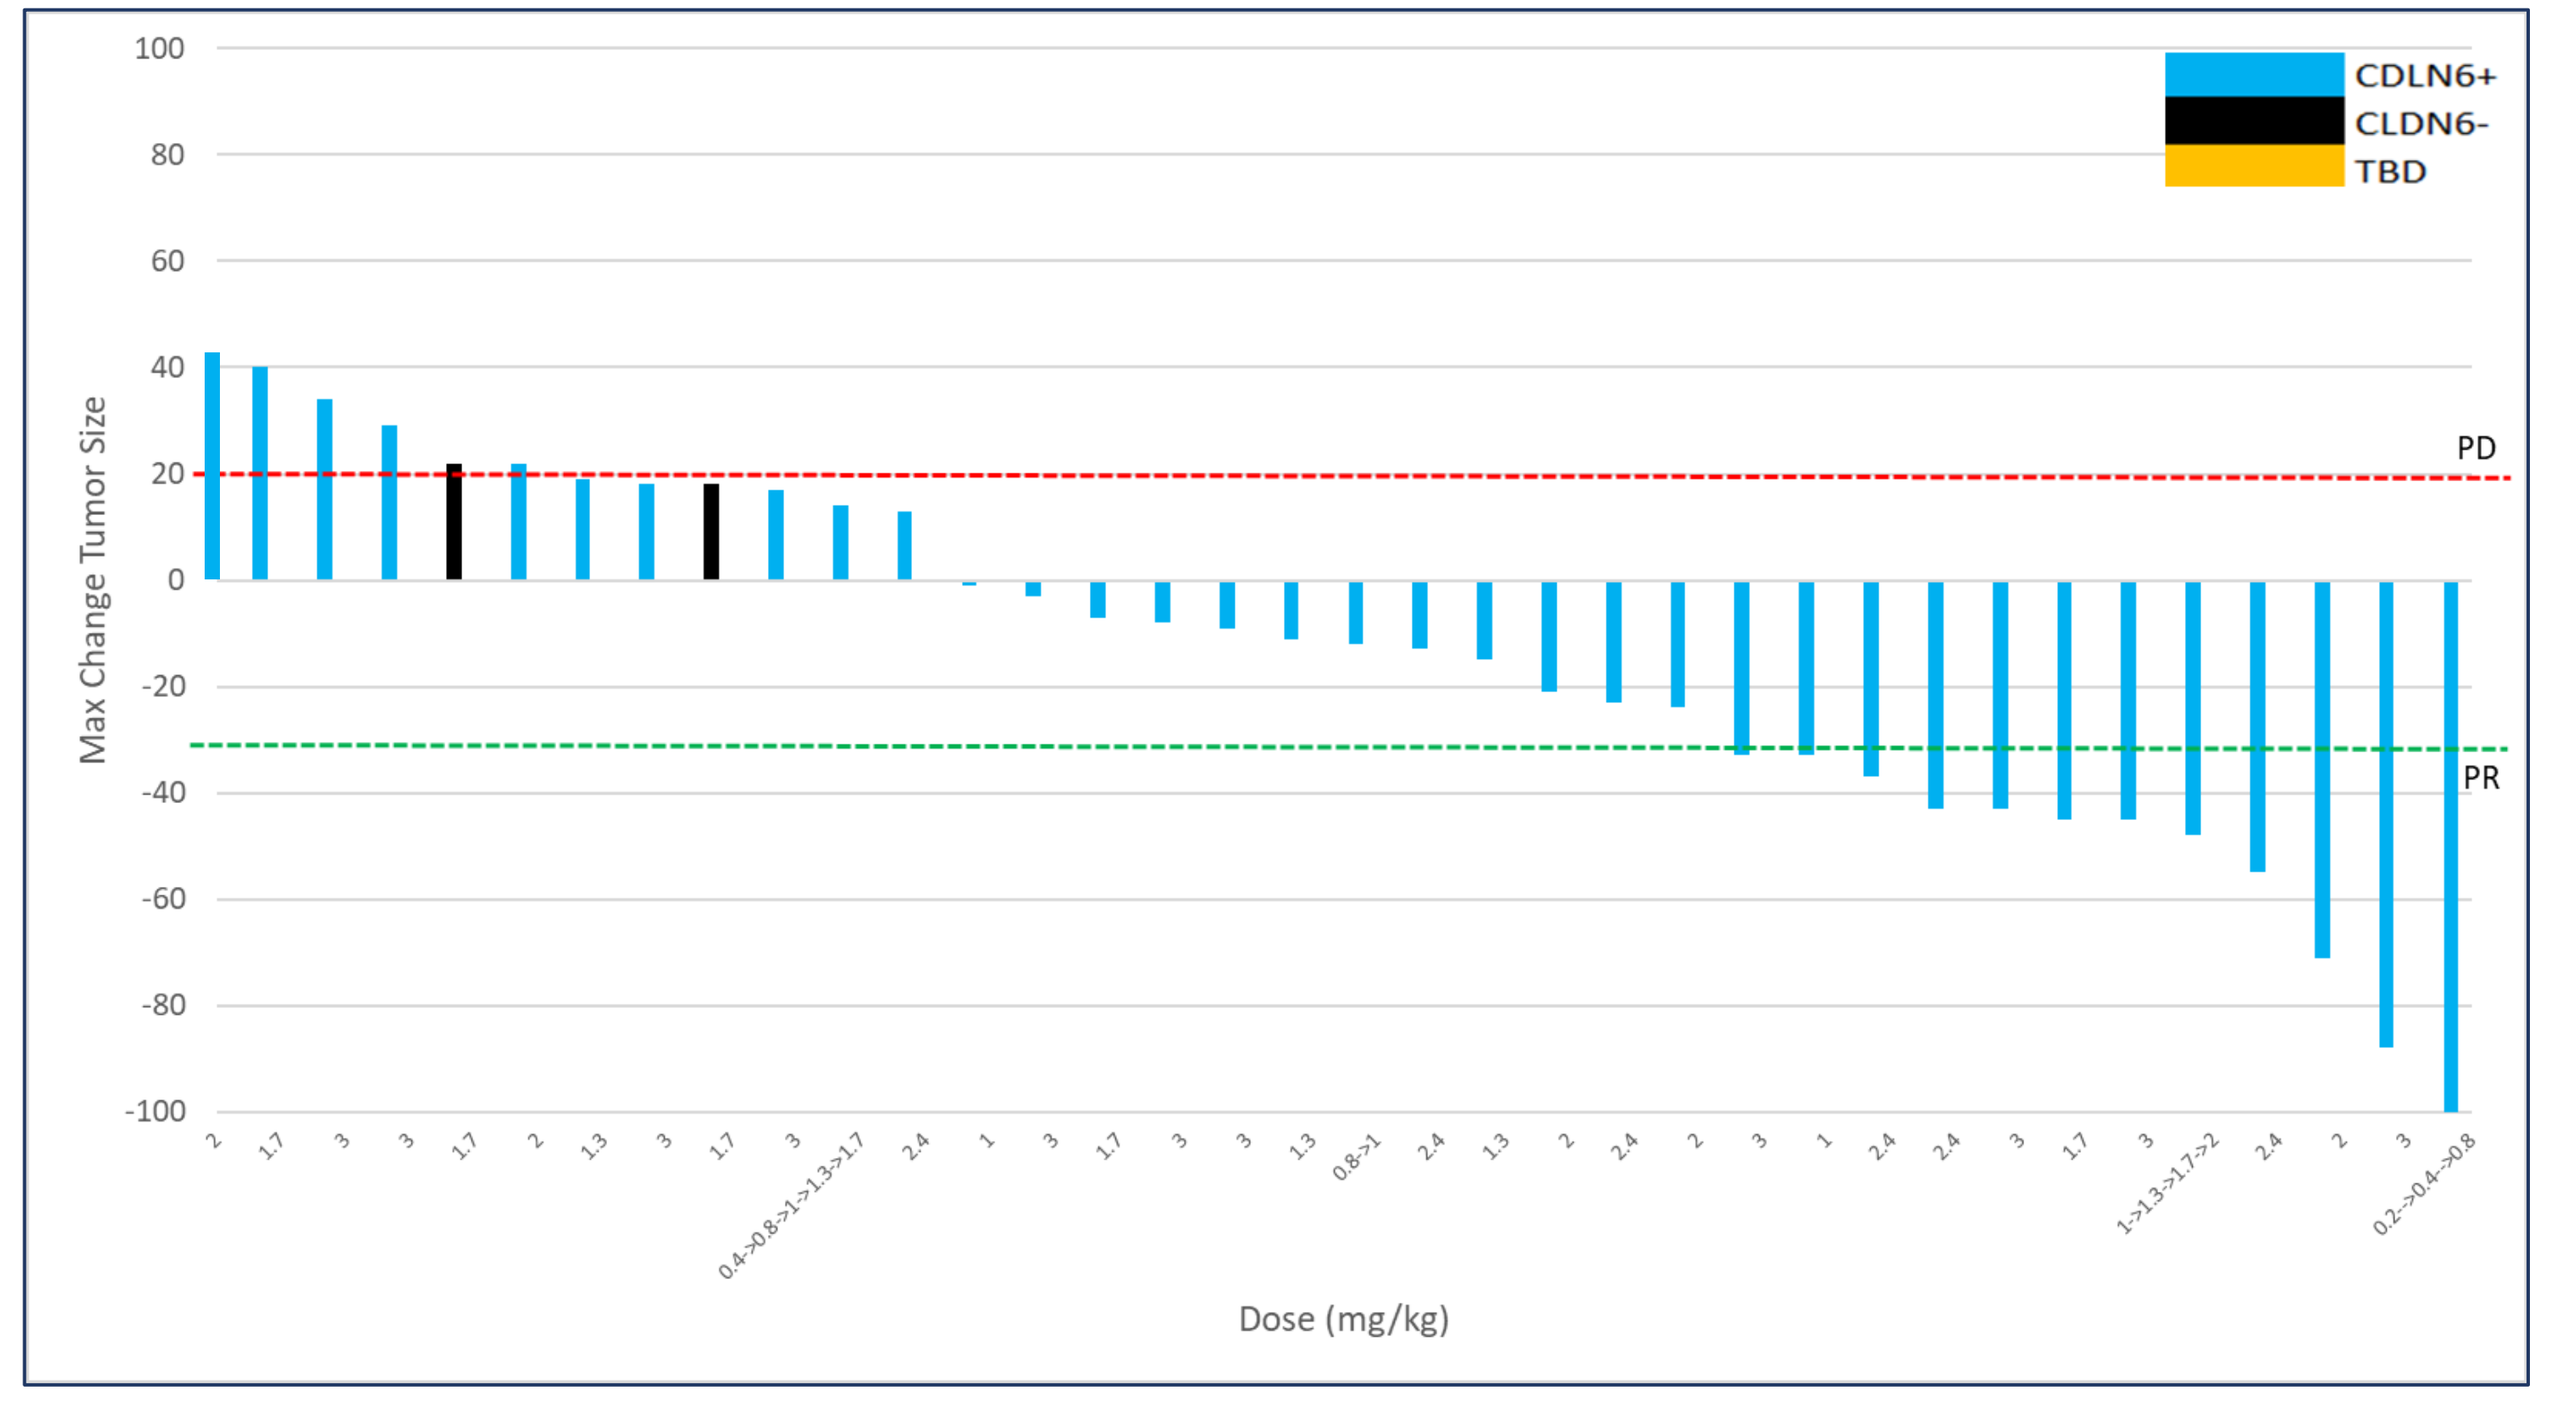
CDLN6+
CLDN6-
TBD
PD
PR
Max Change Tumor Size
Dose (mg/kg)

Datacut 29SEP2023 Dose Levels 0.2 to 3.0 mg/kg (n=36)

Figure 8. Change in Tumor Size Over Time: Dose Levels 0.2 to 3 mg/kg

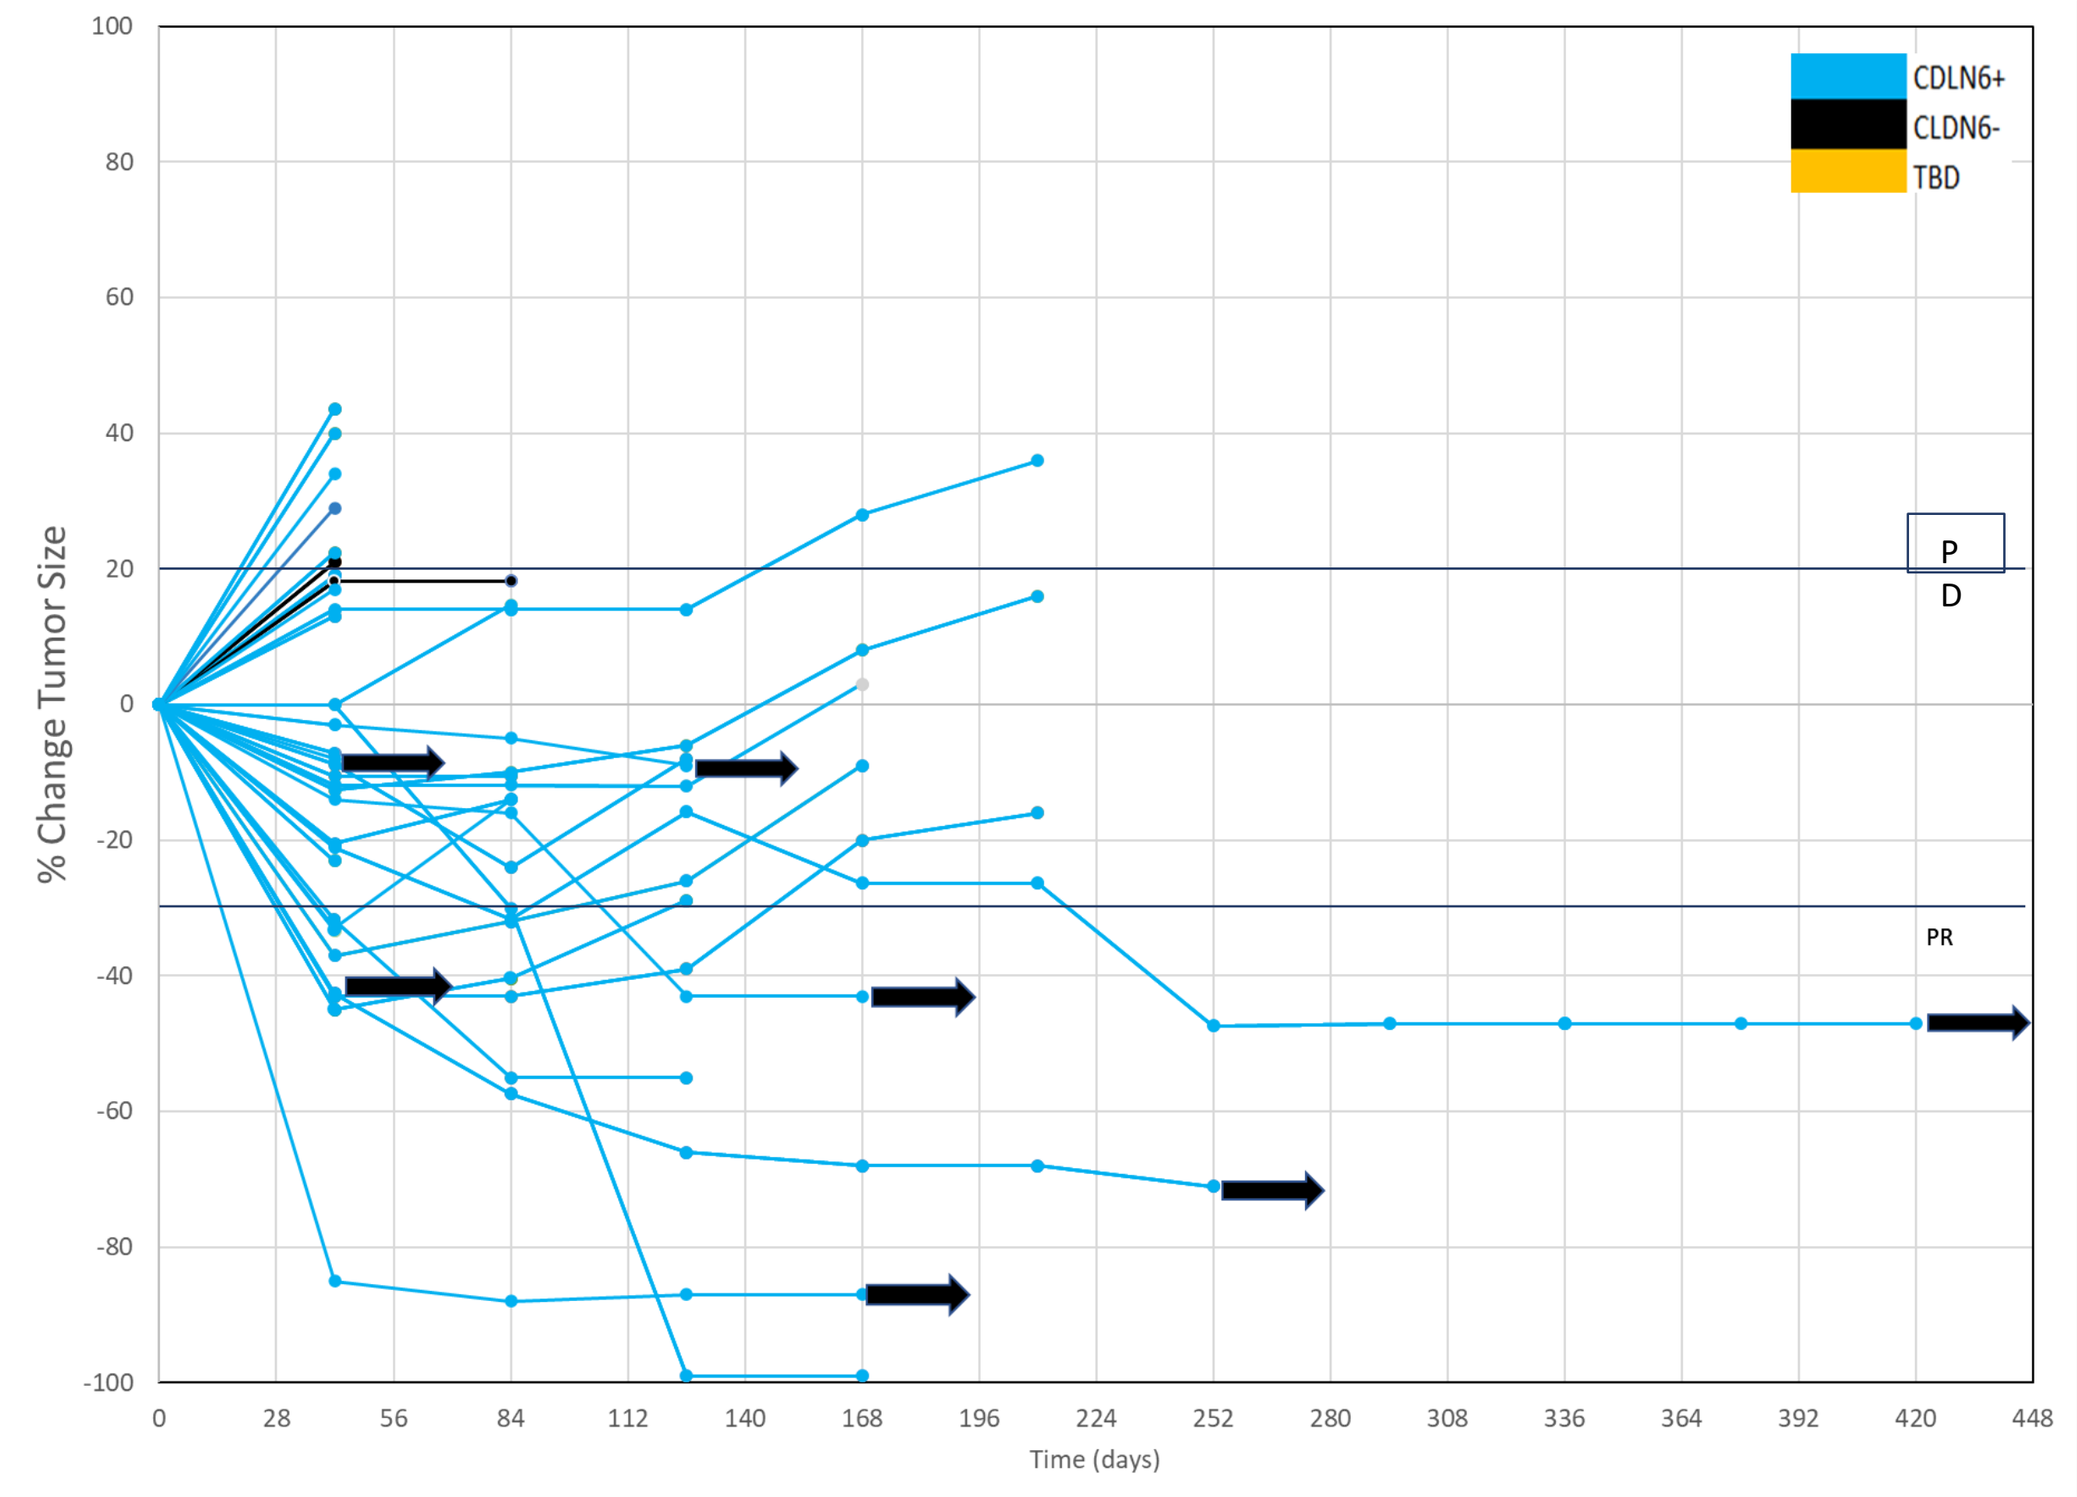
CDLN6+
CLDN6-
TBD
PD
PR
% Change Tumor Size
Time (days)

Datacut 29SEP2023 Dose Levels 0.2 to 3.0 mg/kg (n=36)

Conclusions

- **TORL-1-23 well tolerated from 0.2 to 2.4 mg/kg IV every 21 days**
 - No DLTs 0.2 to 2.4 mg/kg (MTD for other VC-MMAE ADCs)
 - 3.0 mg/kg IV every 21 days
 - G4 neutropenia observed without GCSF
 - No neutropenia with GCSF
- **Initial PK data indicate reduced exposure to free MMAE as compared to other MMAE-containing ADCs**
- **Encouraging antitumor activity observed in heavily pretreated population during dose finding below the MTD**
 - PRs in 11/36 (31%) in evaluable patients across dose levels
 - PRs in 9/27 (33%) with CLDN6+ platinum resistant/refractory ovarian cancer across dose levels
 - PRs in 6/12 (50%) patients across 2.4 mg/kg and 3.0 mg/kg doses (3 additional patients to be evaluated at 3.0 mg/kg)
 - Treatment duration at 2.4 mg/kg is 5 mos and duration at 3.0 mg/kg pending
 - 4.0 mg/kg with GCSF will be explored
- **Dose optimization evaluation to occur in expansion cohorts**

Acknowledgments

- The patients, families, and clinical research colleagues who made this study possible.
- All authors contributed to and approved the presentation.
- Relationships to disclose (G.E. Konecny): Speaker Bureaus for AstraZeneca, GSK, Immunogen, Merck, and research funding to the institution from Lilly and Merck

Correspondence: gkonecny@mednet.ucla.edu

Presented at the European Society of Medical Oncology (ESMO) Annual Meeting 2023, October 20-24, Madrid, Spain & Online

Copies of this poster obtained through (QR) Quick Response and/or text key codes are for personal use only and may not be reproduced without written permission of this authors.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com