175206_poster_big_1.jpg (JPEG Image, 6000 × 3000 pixels) - Sc... https://media4.asco.org/239/13725/175206/175206_poster_big_1.jpg

DPX-Survivac and intermittent low dose cyclophosphamide (CPA) with or without epacadostat (E) in the treatment of subjects with advanced recurrent epithelial ovarian cancer (DeCidE[1] trial): T cell responses and tumor infiltration correlate with tumor regression

Janos L. Tanyi[1], Oliver Dorigo[2], Amit Oza[3], James Strauss[4], Tanya Pejovic[5], Sharad Ghamande[6], Prafull Ghatage[7], Jeannine Villella[8], Stephan Fiset[9], Lisa D. MacDonald[9], Olga Hrytsenko[9], Marianne M. Stanford[9], Robert C. Newton[10], Lance Leopold[10], Gabriela Rosu[9]

[1]University of Pennsylvania, Pennsylvania, USA, [2]Stanford University, California, USA, [3]Princess Margaret Cancer Centre, Ontario, Canada, [4]Mary Crowley Research Center, Texas USA, [5]Oregon Health & Science University, Oregon, USA, [6]Georgia Cancer Center, Georgia, USA, [7]Tom Baker Cancer Centre, Alberta, Canada, [8]Lenox Hill Hospital, New York, USA, [9]IMV Inc., Nova Scotia, Canada, [10]Incyte Corporation, Delaware, USA

2019 ASCO ANNUAL MEETING
CARING FOR EVERY PATIENT, LEARNING FROM EVERY PATIENT
ABSTRACT #5576

Background

Every year, ovarian cancer afflicts approximately 22,000 women in the US and approximately 14,000 will die from the disease. Primary treatment consists of debulking surgery followed by systemic chemotherapy with or without the addition of targeted therapies. Despite achieving initial benefits, a significant number of these women will experience recurrence after their 1st or 2nd line therapy (1L or 2L).

The introduction of immunotherapies and their known success in several indications was thought to bring a revolution for the treatment of ovarian cancer; unfortunately this revolution has yet to be realized. There remains a significant unmet medical need for novel therapies that will help ovarian cancer patients live better and longer. Consequently, there is also a pressing need to identify biomarkers or clinical features that are predictive of response, thus providing substantial benefits to the patients.

DPX-Survivac is a novel and unique T cell activation therapy that generates *de novo* T cells against survivin. In maintenance Phase 1 and 1b studies in ovarian cancer, it was shown that DPX-Survivac can generate a strong and specific T cell response against survivin expressed on the surface of the tumor and long progression free interval have been observed in subjects (> 7 years). DPX-Survivac showed a well-tolerated safety profile with majority of events being grade 1 and 2 local site reactions.

Intermittent low dose cyclophosphamide (CPA) is used as a "biological response modifier". Studies have shown that low does of CPA can have selective effects on the immune system that may augment the efficacy of immunotherapies.

Epacadostat (E) is an IDO-1 inhibitor that prevents the oxidation of tryptophan into kynurenine. This inhibition has been shown to increase and restore the proliferation and activation of immune cells.

Study Design

- Multicenter Phase 1b/2 study, ongoing in the United States and Canada
- Primary objectives: safety, cell mediated immunity, changes in immune cell infiltration of tumor
- Secondary objectives: ORR, DCR, DoR using RECIST v1.1, TTP, OS, and CA-125 response & progression
- DPX-Survivac is administered subcutaneously 2 x 0.25 mL q3w followed by up to 6 x 0.1 mL q8w; oral CPA 50 mg BID on alternating weeks; oral epacadostat (E) up to 300 mg BID

Phase 1: dosing finding, non randomized, N = 53

DPX-Survivac, CPA, 100 mg E → assess DLT → DPX-Survivac, CPA, 300 mg E → assess DLT → DPX-Survivac, CPA, RP2D E

Phase 2: 1:1 randomization with interim analysis

DPX-Survivac, CPA, RP2D E / DPX-Survivac, CPA → Review of emerging data / interim analysis → **Enrollment ongoing** DPX-Survivac, CPA

Demographics

Table 1: Baseline subject demographics and disposition (N=67)

Parameter	Statistic	All n (%)	With Epacadostat (E) Phase 1b (N=53)	With Epacadostat (E) Phase 2 (N=7)	No E Phase 2 (N=7)
Age (years)	Median	65	66	64	58
	Min, max	35, 87	35, 87	58, 78	35, 77
Race n (%)	White	60 (89.6)	47 (88.7)	7 (100)	6 (85.7)
	Black or African American	3 (4.5)	3 (5.7)	0 (0)	0 (0)
	Asian	1 (1.5)	0 (0)	0 (0)	1 (14.3)
	Other	3 (4.5)	3 (5.7)	0 (0)	0 (0)
ECOG	0	38 (56.7)	29 (54.7)	5 (71.4)	4 (57.1)
	1	29 (43.3)	24 (45.3)	2 (28.6)	3 (42.9)
Cancer type	Epithelial ovarian	51 (76.1)	39 (73.6)	6 (85.7)	6 (85.7)
	Fallopian tube	6 (9.0)	6 (11.3)	0 (0)	0 (0)
	Peritoneal	10 (14.9)	8 (15.1)	1 (14.3)	1 (14.3)
Number of previous lines of treatment	1	9 (13.4)	8 (15.1)	0 (0)	1 (14.3)
	2	12 (17.9)	11 (20.8)	1 (14.3)	0 (0)
	3	9 (13.4)	8 (15.1)	0 (0)	1 (14.3)
	4	13 (19.4)	10 (18.9)	0 (0)	3
	≥ 5	24 (35.8)	16 (30.2)	6 (85.7)	2 (28.6)
Sensitivity to most recent platinum	Sensitive	19 (28.4)	18 (34.0)	0 (0)	1 (14.3)
	Resistant/ refractory	48 (71.6)	35 (66.0)	7 (100)	6 (85.7)
Best response to most recent platinum	Unknown	14 (20.9)	8 (15.1)	3 (42.9)	3 (42.9)
	Progressive disease	12 (17.9)	8 (15.1)	3 (42.9)	1 (14.3)
	Stable disease	12 (17.9)	11 (20.8)	0 (0)	1 (14.3)
	Partial response	14 (20.9)	11 (20.8)	1 (14.3)	2 (28.6)
	Complete response	15 (22.3)	15 (28.3)	0 (0)	0 (0)
Evaluable	D56 scan/biopsy complete	40 (59.7)	32 (60.4)	4 (57.1)	4 (57.1)
Early withdrawal	Disease progression	33 (49.3)	27 (50.9)	4 (57.1)	2 (28.6)
	Adverse event	12 (17.9)	12 (22.6)	0 (0)	0 (0)
	Withdrawal of consent	8 (11.9)	5 (9.4)	0 (0)	3 (42.9)
	Investigator judgement	4 (6.0)	4 (7.5)	0 (0)	0 (0)
	Other	2 (3.0)	1 (1.9)	1 (14.3)	0 (0)

Safety

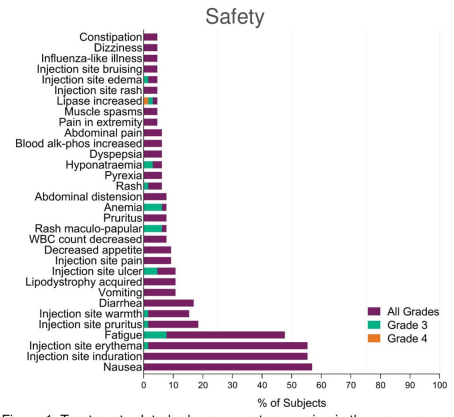
Constipation
Dizziness
Influenza-like illness
Injection site bruising
Injection site edema
Injection site rash
Lipase increased
Muscle spasms
Pain in extremity
Abdominal pain
Blood alk-phos increased
Dyspepsia
Hyponatraemia
Pyrexia
Rash
Abdominal distension
Anemia
Pruritus
Rash maculo-papular
WBC count decreased
Decreased appetite
Injection site pain
Injection site ulcer
Lipodystrophy acquired
Vomiting
Diarrhea
Injection site warmth
Injection site pruritus
Fatigue
Injection site erythema
Injection site induration
Nausea
All Grades
Grade 3
Grade 4
% of Subjects

Figure 1: Treatment-related adverse events occurring in three or more subjects (5%). AE are counted once per subject at the highest grade observed. N=60 for DPX-Survivac and CPA with E; N=5 without E.

Long Lasting Responses

- Earlier studies with DPX-Survivac have shown durable immune responses and a possible improvement to PFS
- In the 1L or 2L maintenance setting, some subjects gained an additional 48 months or more PFS following treatment with DPX-Survivac and CPA
- In this study, most subjects had 4 or more lines of prior therapy
- 3/53 Phase 1b subjects completed a full year of study treatment with two electing to continue with long term treatment

Table 2: Comparison of time to progression for subjects who achieved a partial response (PR) in the Phase 1b

Subject ID	Prior Lines	Response to Last Platinum	Time to Progression - Last Therapy	Time to Progression - DeCidE[1] Study
0601	5	Sensitive	Progressed on treatment: 4.5 months	21.5 months
0606	1	Sensitive	15.6 months	> 25.6 months
0611	2	Sensitive	32.5 months	Progressed on treatment: 4.6 months
0614	1	Resistant	9.9 months	> 21.2 months
0635	2	Refractory	Progressed on treatment: 2.1 months	Progressed on treatment: 4.7 months

Case Study (Figures 2 - 6)

55 y, fallopian tube cancer, platinum resistant, 1 previous line of therapy, tumor burden 20.3 mm, ECOG = 1, HLA-A01
DPX-Survivac + CPA + 300 mg BID epacadostat

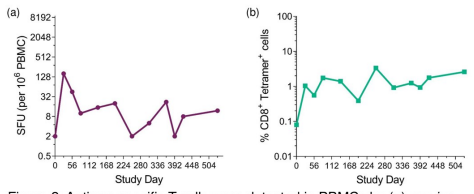
(a)
SFU (per 10^6 PBMC)
Study Day
(b)
% CD8+ Tetramer+ cells
Study Day

Figure 2: Antigen-specific T cells were detected in PBMCs by (a) *ex vivo* IFN-γ ELISPOT with survivin peptide stimulation and (b) MHC multimer staining using the HLA-A01 matched survivin peptide.

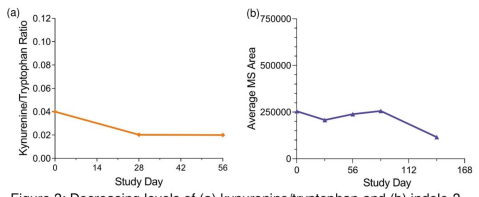
(a)
Kynurenine/Tryptophan Ratio
Study Day
(b)
Average MS Area
Study Day

Figure 3: Decreasing levels of (a) kynurenine/tryptophan and (b) indole-3-acrylic acid (by untargeted metabolomics assessment) were measured in the blood following study treatment.

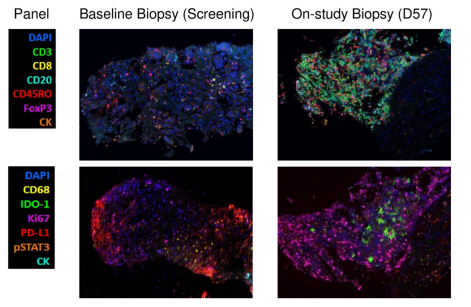
Panel
Baseline Biopsy (Screening)
On-study Biopsy (D57)
DAPI
CD3
CD8
CD20
CD45RO
FoxP3
CK
DAPI
CD68
IDO-1
Ki67
PD-L1
pSTAT3
CK

Figure 4: Increases in tumor immune infiltration demonstrated by multiplex IHC. A pre-treatment biopsy was taken before study enrollment and an on-treatment biopsy between D56 and D70. Unmixed composite fluorescence IHC images are shown (Analysis was conducted by PerkinElmer.)

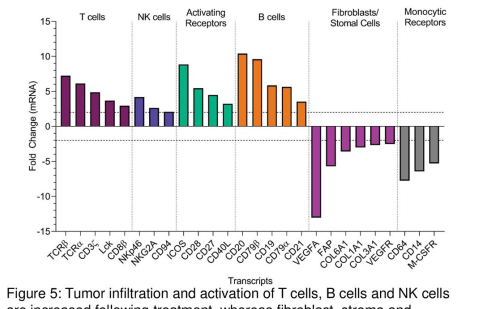
T cells
NK cells
Activating Receptors
B cells
Fibroblasts/ Stomal Cells
Monocytic Receptors
Fold Change (mRNA)
Transcripts

Figure 5: Tumor infiltration and activation of T cells, B cells and NK cells are increased following treatment, whereas fibroblast, stroma and monocytic markers are substantially reduced by treatment. Analysis is performed at the RNA level using whole exome sequencing of tumor biopsy samples. Fold-changes in the expression of each transcript were calculated relative to baseline levels. (Sequencing and data normalization were conducted by Q[2] Solutions)

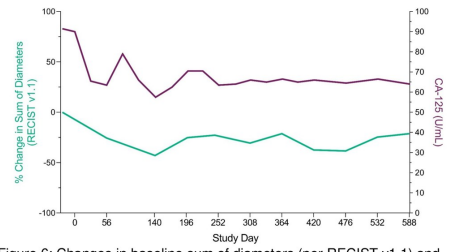
% Change in Sum of Diameters (RECIST v1.1)
CA-125 (U/mL)
Study Day

Figure 6: Changes in baseline sum of diameters (per RECIST v1.1) and levels of CA-125 following study treatment

Randomized Phase 2

Table 3: Best Response per RECIST v1.1 criteria observed in subjects randomized to treatment of DPX-Survivac and CPA with or without epacadostat

Subject ID	+ or - Epacadostat	Best Response
2602	+	N/E
2604	+	PD
2606	+	PD
2607	+	PD
2609	+	SD
2611	+	PD
2612	+	PD

Subject ID	+ or - Epacadostat	Best Response
2601	-	SD
2603	-	SD
2605	-	PD
2608	-	SD
2610	-	SD

N/E = not evaluable, subject withdrawn before dosing

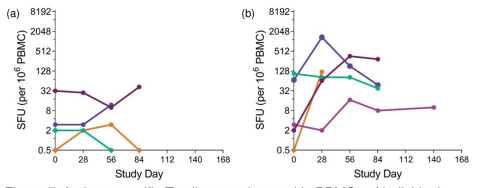
(a)
SFU (per 10^6 PBMC)
Study Day
(b)
SFU (per 10^6 PBMC)
Study Day

Figure 7: Antigen-specific T cells were detected in PBMCs of individual subjects by *ex vivo* IFN-γ ELISPOT with survivin peptide stimulation. Subjects randomized to treatment (a) with epacadostat and (b) without.

Conclusions

- 67 recurrent ovarian cancer subjects were enrolled to receive DPX-Survivac and low dose cyclophosphamide with up to 300 mg BID epacadostat (N=60) or without (N=7)
- Treatments have been well tolerated
- Robust systemic survivin-specific T cell responses
- Infiltration of T cells into tumors post treatment correlates with durable clinical responses
- Responders have shown prolonged duration of clinical benefits reaching up to more than 2 years, increasing the duration of clinical benefits to previous line of chemotherapy
- Phase 2 study is ongoing, treating with DPX-Survivac and low dose cyclophosphamide

Further Information

Corresponding Author: grosu@imv-inc.com
ClinicalTrials.gov Identifier: NCT02785250
Sponsored by IMV Inc. with co-funding from Incyte Corporation
Copies of this poster obtained through QR are for personal use only and my not be reproduced without permission from ASCO and the authors

1 of 1 2/27/20, 10:26 AM