Phase Ib study of adavosertib in combination with olaparib in patients with refractory solid tumors: dose escalation

Erika Hamilton,[1] Gerald S Falchook,[2] Judy S Wang,[1,3] Siqing Fu,[4] Amit M Oza,[5] Karen So,[6] Esteban Rodrigo Imedio,[6] Sanjeev Kumar,[6] Lone Ottesen,[6] Ganesh M Mugundu,[7] Juliann Chmielecki,[8] Suzanne Jones,[1] David R Spigel,[1] Bob T Li[9]

[1]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA; [2]Sarah Cannon Research Institute at HealthONE, Denver, CO, USA; [3]Florida Cancer Specialists, Sarasota, FL, USA; [4]MD Anderson Cancer Center, Houston, TX, USA; [5]Princess Margaret Cancer Centre, Toronto, Canada; [6]AstraZeneca, Cambridge, UK; [7]Quantitative Clinical Pharmacology, Early Clinical Development, IMED Biotech Unit, AstraZeneca, Boston, MA, USA; [8]Translational Science, Oncology, IMED Biotech Unit, AstraZeneca, Waltham, MA, USA; [9]Memorial Sloan Kettering Cancer Center, New York, NY, USA

CT025

Introduction

- Adavosertib (AZD1775) is a highly selective small-molecule WEE1 inhibitor being developed as a treatment for advanced solid tumors and has shown single-agent activity in patients with BRCA1/2 mutations (BRCAm).[1]
- Preclinical data suggest that adavosertib enhances the antitumor effect of PARP inhibitors such as olaparib[2,3]
 - Olaparib has demonstrated clinical activity in ovarian, breast, prostate and pancreatic cancers[4,5]
 - Enhanced tumor regression has been seen with adavosertib plus olaparib in patient-derived xenograft (PDX) models of ovarian, triple-negative breast and small-cell lung cancer.[6]
- This Phase Ib study (NCT02511795, D6010C00005, REFMAL384) investigated the safety and tolerability, and defined the maximum tolerated dose (MTD) and recommended Phase II dose (RP2D), of adavosertib plus olaparib in patients with refractory solid tumors. Here, we report the initial dose-escalation part of this study.

Study objectives

- Primary: determine MTD/RP2D of adavosertib combined with olaparib in patients with refractory solid tumors.
- Secondary: investigate the safety and tolerability, clinical activity and pharmacokinetic (PK) profile of adavosertib combined with olaparib.

Methods

- Key inclusion criteria were: age ≥18 years; Eastern Cooperative Oncology Group (ECOG) performance status 0–1; radiation therapy completed ≥7 days before study treatment; histologically confirmed refractory solid tumor for which there is no established treatment with curative intent; and ≥1 prior course of systemic therapy.
- Patients received adavosertib (once daily [qd] or twice daily [bid]) for 3 consecutive days with 4 days off treatment (3/4), or 5 consecutive days with 2 days off (5/2; qd cohorts only), plus continuous oral olaparib (bid) for 14 or 21 days of a 21-day cycle.
- The MTD was the highest dose at which less than one-third of evaluable patients had a dose-limiting toxicity (DLT), defined during cycle 1 as:
 - Hematologic grade ≥4 adverse events (AEs) lasting >7 days
 - Grade 3 thrombocytopenia with grade ≥2 bleeding
 - Non-hematologic grade ≥3 AEs (excluding nausea, vomiting or diarrhea that responds to supportive care)
 - Liver function tests (LFTs): grade ≥3 total bilirubin, increased hepatic alanine aminotransferase, aspartate aminotransferase or alkaline phosphatase lasting >48 hours, or any change in LFTs consistent with Hy's Law
 - Any other toxicity that disrupted dosing for >7 days or was judged to be a DLT.
- Clinical activity was assessed in terms of objective response rate (ORR), progression-free survival (PFS) and disease control rate (DCR).
- The PK profile of adavosertib in combination with olaparib was evaluated throughout the study
 - Pre-adavosertib PK values for olaparib were determined via a substudy in which single-agent olaparib was given bid, orally, for 3 consecutive days, as determined by the adavosertib cohort assignment.
- Blood and tissue samples were collected at specified intervals for biomarker analyses.
- All participants provided written informed consent, and the study was conducted in accordance with the declaration of Helsinki.

Results

Study participants

- A total of 119 patients were treated (Table 1)
 - More than two-thirds of participating patients were female (70.6%), and the median age at study entry was 59 years
 - The most common primary tumor sites were ovary (21.0%), breast (16.0%) and lung (12.6%).

Table 1. Patient and disease characteristics at baseline

	Total population (N=119)
Age, years	
Mean (SD)	58.9 (10.7)
Median (range)	59 (26–80)
Male/female, %/%	29.4/70.6
Race, n (%)	
White	105 (88.2)
Black/African American	6 (5.0)
Asian	5 (4.2)
Other	3 (2.5)
Mean body mass index, kg/m² (SD)	27.3 (6.6)
ECOG performance status, n (%)	
0 (normal activity)	36 (30.3)
1 (symptoms but ambulatory)	83 (69.7)
Primary diagnosis, n (%)	
Ovary	25 (21.0)
Breast	19 (16.0)
Lung	15 (12.6)
Prostate	9 (7.6)
Uterus	9 (7.6)
Colon	6 (5.0)
Pancreas	6 (5.0)
Rectal	4 (3.4)
Soft tissue	3 (2.5)
Central nervous system	1 (0.8)
Fallopian tube	1 (0.8)
Liver	1 (0.8)
Other	20 (16.8)
Mean number of previous therapy regimens (SD)	
Systemic	4.1 (2.5)
Radiotherapy	1.1 (1.5)
Disease-related surgery	2.3 (2.5)

SD, standard deviation

Tolerability

- Overall, DLTs were reported in 14 (14.3%) of 98 evaluable patients (Table 2)
 - The most common DLTs were thrombocytopenia (n=4; two grade 3 and two grade 4) and neutropenia (n=4; one grade 2, one grade 3 and two grade 4); two patients experienced both. No bleeding events were associated with thrombocytopenia
 - The MTD with a bid adavosertib dosing schedule was in cohort 4.2 (1 in 11 patients [9.1%] had a DLT) and with a qd dosing schedule was in cohort 7.4 (1 in 13 patients [8.3%] had a DLT).
- AEs were reported in 118 (99.2%) patients, with grade ≥3 AEs occurring in 73 (61.3%; Table 2)
 - The most common grade ≥3 AEs were anemia (n=28, 23.5%), neutropenia (n=26, 21.8%) and thrombocytopenia (n=20, 16.8%; all grouped terms)
 - Grade ≥3 AEs considered to be causally related to adavosertib or olaparib occurred in 51 (42.9%) and 52 (43.7%) patients, respectively.
- There were four AEs with an outcome of death (sepsis [n=2], respiratory failure and cardiovascular disease [both n=1]); one (sepsis) was considered causally related to adavosertib but was not a result of a DLT.

Clinical response

- In the total population, ORR was 11.1% (Table 3)
 - For the cohorts in which MTD/RP2D for bid dosing (cohort 4.2) and RP2D for qd dosing (cohort 7.4) were determined, ORRs were 30.8% and 0%, respectively.
- In the total population, PFS was 3.2 months (95% confidence interval [CI] 2.46–4.11; Table 3)
 - For cohorts 4.2 and 7.4, median PFS was 7.4 months (95% CI 2.46–15.77) and 1.4 months (95% CI 1.35–4.80), respectively.
- In the total population, DCR was 55.7% (Table 3)
 - For cohorts 4.2 and 7.4, DCRs were 76.9% and 53.8%, respectively.

Table 2. Summary of treatment schedules and tolerability profiles

Cohort	Adavosertib dose (schedule)	Olaparib dose	Adavosertib schedule, days	Olaparib schedule, days	Patients, n (n evaluable*)	Patients with a DLT, n (%)	Grade ≥3 AEs,† n (%)
1	125 mg bid (3/4)	100 mg bid	1–3/8–10	1–14	3 (2)	0	1 (33.3)
2	150 mg bid (3/4)		1–3/8–10	1–14	4 (4)	0	2 (50.0)
3.1	175 mg bid (3/4)		1–3/8–10	1–14	4 (2)	0	3 (75.0)
3.2	150 mg bid (3/4)	200 mg bid	1–3/8–10	1–14	7 (5)	0	4 (57.1)
4.1	175 mg bid (3/4)		1–3/8–10	1–14	7 (7)	0	4 (57.1)
4.2	175 mg bid (3/4)		1–3/8–10	1–21	14 (11)	1 (9.1)	9 (64.3)
4.3	175 mg bid (3/4)		1–3/8–10/15–17	1–21	14 (11)	2 (18.2)	13 (92.9)
5	175 mg bid (3/4)	300 mg bid	1–3/8–10	1–14	5 (5)	1 (20.0)	3 (60.0)
6.1	250 mg qd (5/2)	200 mg bid	1–5/8–12	1–21	7 (4)	2 (50.0)	6 (85.7)
6.2	**200 mg qd (5/2)**		**1–5/8–12**	**1–21**	**7 (5)**	**2 (40.0)**	**4 (57.1)**
7.1	250 mg qd (3/4)		1–3/8–10	1–21	16 (14)	2 (14.3)	12 (75.0)
7.2	250 mg qd (3/4)		1–3/8–10/15–17	1–21	4 (4)	1 (25.0)	3 (75.0)
7.3	300 mg qd (3/4)		1–3/8–10	1–21	3 (3)	1 (33.3)	2 (66.7)
7.4	200 mg qd (3/4)		1–3/8–10	1–21	13 (12)	1 (8.3)	3 (23.1)
8.1	200 mg qd (3/4)	300 mg bid	1–3/8–10	1–21	11 (9)	1 (11.1)	4 (36.4)

Bold indicates the RP2D (adavosertib 200 mg qd plus olaparib 200 mg bid) being carried forward into the dose-expansion part of this study. *Evaluable patients received >75% of the planned dose of adavosertib and olaparib; †Common Terminology Criteria for Adverse Events

Table 3. Summary of response, by cohort

Cohort	Patients, n	Evaluable for ORR, n*	ORR, n (%)	Median PFS, months	Evaluable for DCR, n*	DCR, n (%)
1	3	3	0	5.6	3	3 (100)
2	4	4	1 (25.0)	2.9	4	2 (50.0)
3.1	4	3	0	6.0	4	3 (75.0)
3.2	7	7	1 (14.3)	10.9	7	5 (71.4)
4.1	7	7	1 (14.3)	2.6	7	4 (57.1)
4.2	14	13	4 (30.8)	7.4	13	10 (76.9)
4.3	14	13	1 (7.7)	2.7	14	7 (50.0)
5	5	4	2 (50.0)	9.8	4	4 (100)
6.1	7	5	1 (20.0)	1.4	6	1 (16.7)
6.2	7	7	0	1.3	7	1 (14.3)
7.1	16	15	0	4.0	16	10 (62.5)
7.2	4	4	1 (25.0)	NC	4	3 (75.0)
7.3	3	3	0	1.4	3	0
7.4	13	11	0	1.4	13	7 (53.8)
8.1	11	9	0	NC	10	4 (40.0)

*For ORR and DCR, evaluable patients have measurable disease and received >75% of the planned dose of adavosertib and olaparib. NC, not calculated

Pharmacokinetics

- Following dosing on cycle 1 day 3, adavosertib 175 mg bid plus olaparib 200 mg bid had the greatest cover during the dosing interval over pCDK1 IC_{50} and preclinical cell kill activity (Figure 1).
- Drug–drug interaction studies showed that adavosertib 175 mg bid increased olaparib area under the concentration–time curve (AUC) by 33% and maximum plasma concentration (C_{max}) by 18% (Figure 2 and Table 4).

Figure 1. Comparison of PK across adavosertib doses in combination with olaparib 200 mg bid

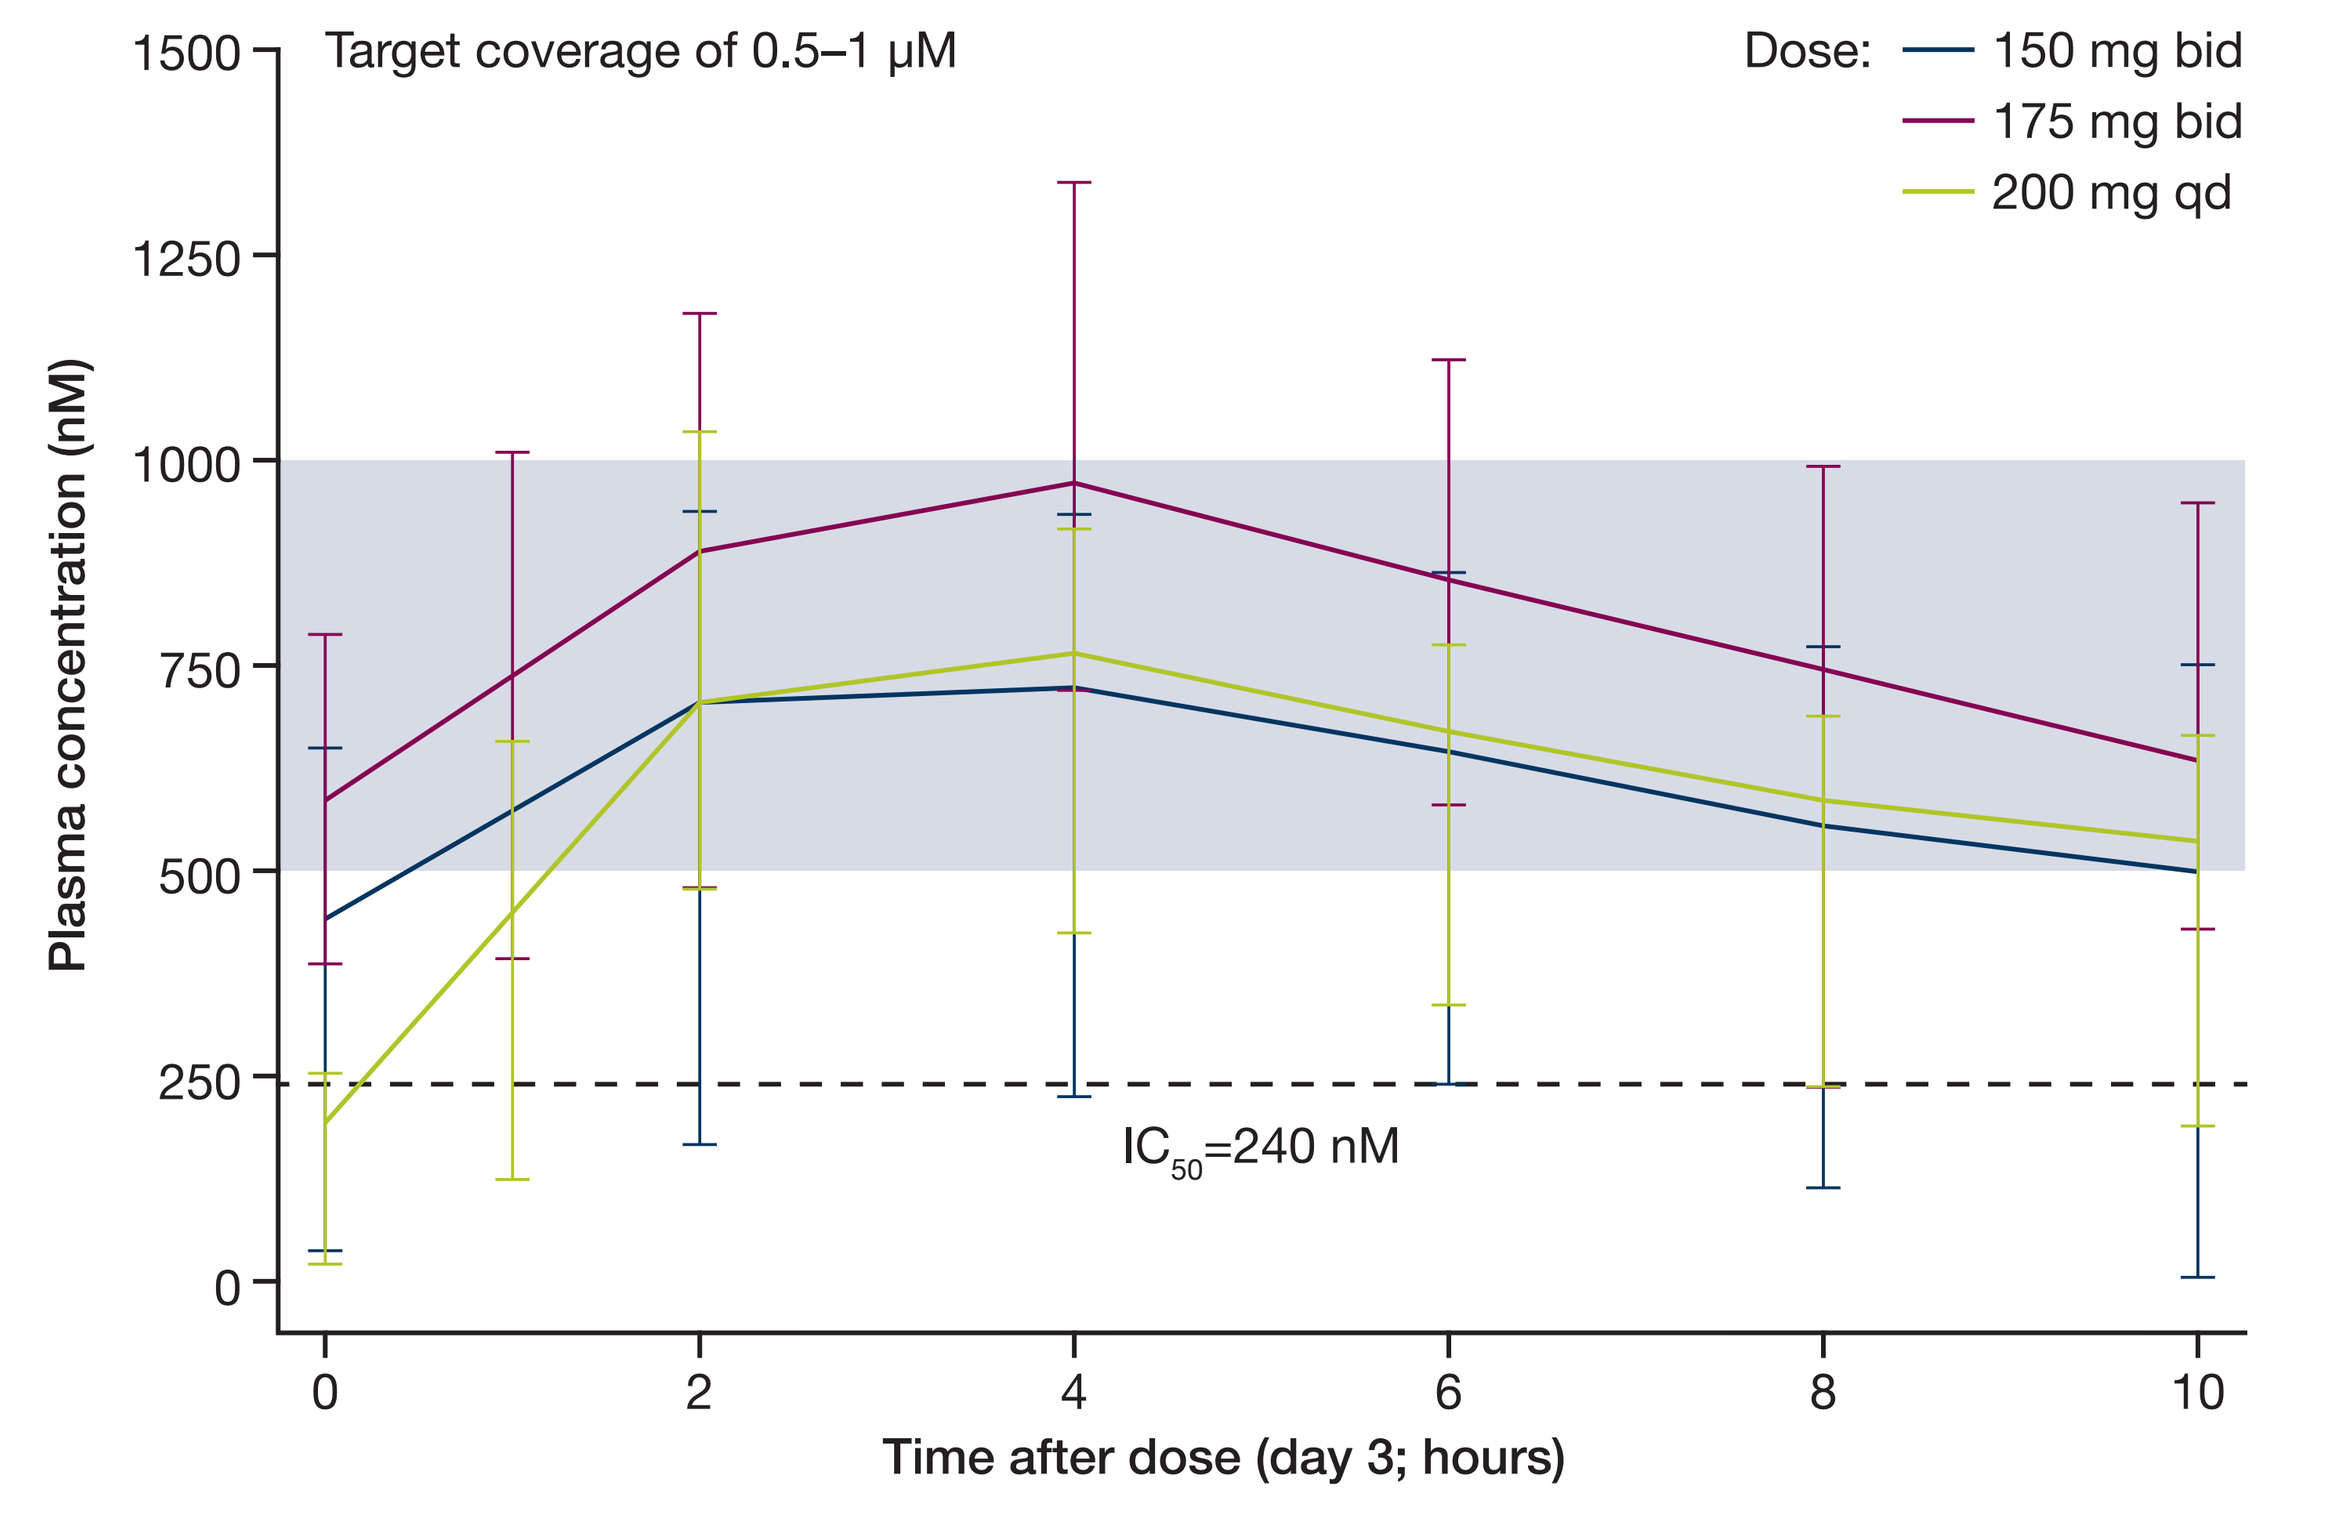
Target coverage of 0.5–1 µM
Dose:
150 mg bid
175 mg bid
200 mg qd
Plasma concentration (nM)
1500
1250
1000
750
500
250
0
IC50=240 nM
0
2
4
6
8
10
Time after dose (day 3; hours)

Data are shown as geometric mean ± SD. IC_{50}, 50% inhibitory concentration

Figure 2. Olaparib clinical $C_{min,ss}$ compared with preclinical PDX PAR IC_{95}

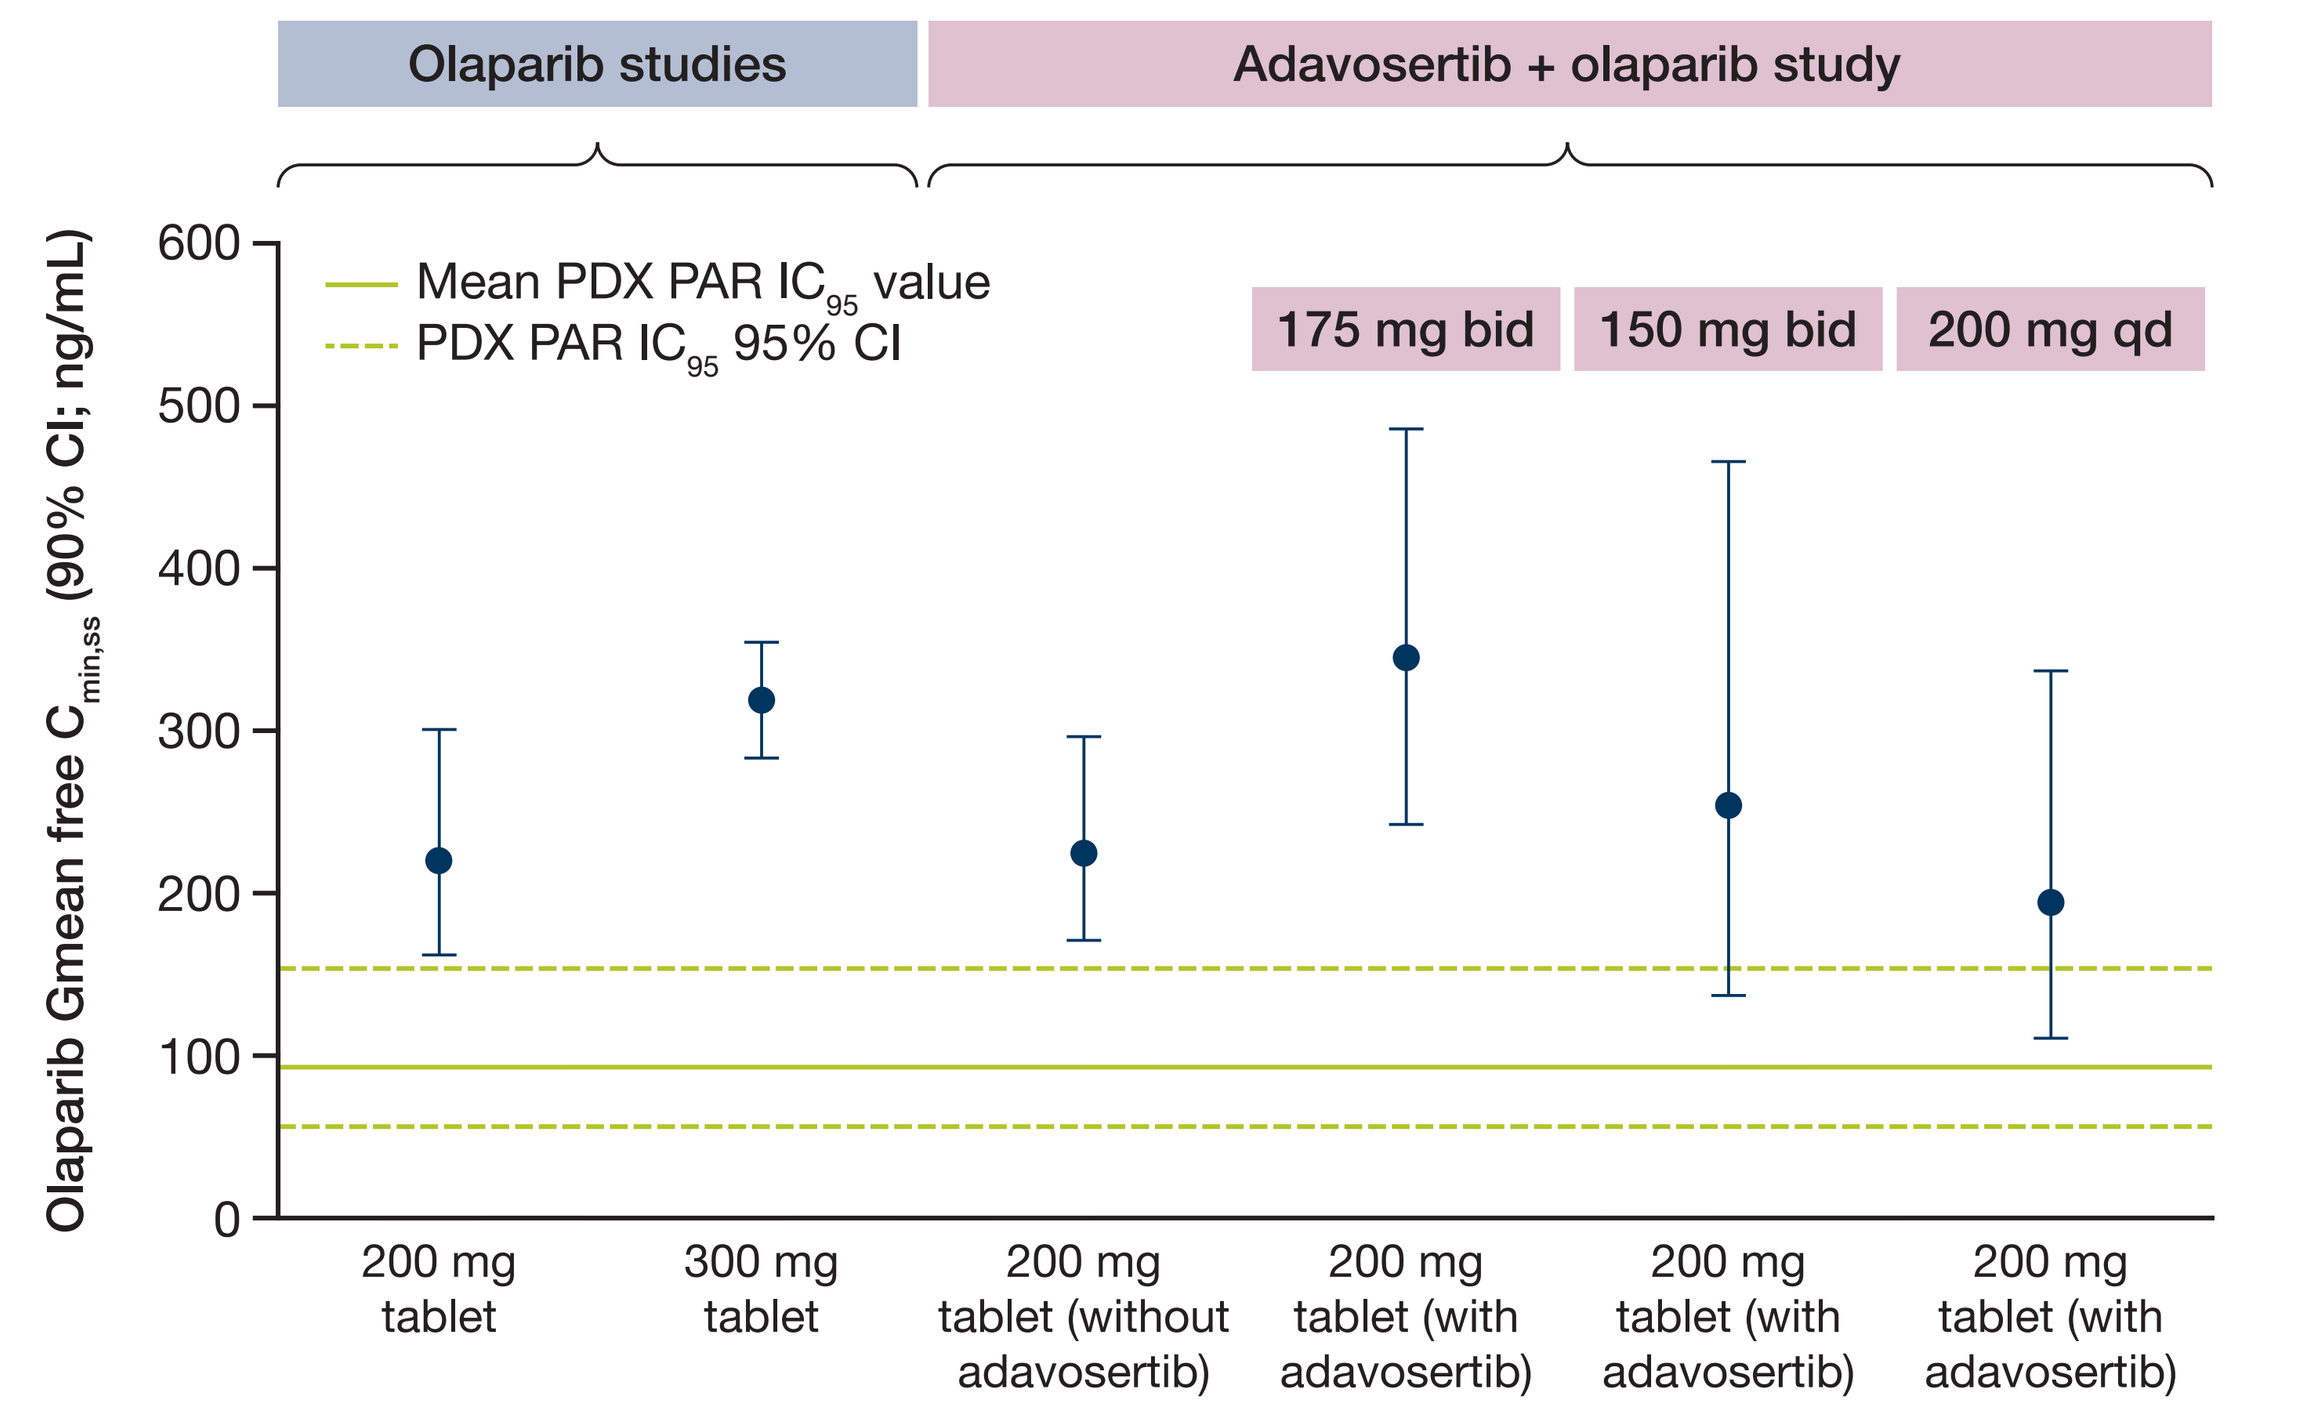
Olaparib studies
Adavosertib + olaparib study
Mean PDX PAR IC95 value
PDX PAR IC95 95% CI
175 mg bid
150 mg bid
200 mg qd
Olaparib Gmean free Cmin,ss (90% CI; ng/mL)
600
500
400
300
200
100
0
200 mg tablet
300 mg tablet
200 mg tablet (without adavosertib)
200 mg tablet (with adavosertib)
200 mg tablet (with adavosertib)
200 mg tablet (with adavosertib)

$C_{min,ss}$, minimum plasma concentration at steady state; IC_{95}, 95% inhibitory concentration; PAR, protease-activated receptor

Table 4. Changes in olaparib PK following combination with adavosertib

Adavosertib dose (schedule)	Olaparib dose	N	$AUC_{0–10}$ ratio (range)	C_{max} ratio (range)
175 mg bid (3/4)	200 mg bid	23	1.33 (0.74–2.57)	1.18 (0.85–2.11)
150 mg bid (3/4)	200 mg bid	6	0.89 (0.50–1.44)	0.91 (0.42–1.38)
200 mg qd (3/4)	200 mg bid	11	1.08 (0.80–2.05)	1.02 (0.62–1.48)

$AUC_{0–10}$, AUC from time 0 to 10 hours

Biomarkers

- Preliminary analyses of samples showed that *TP53* mutation was the most common genetic aberration, found in 71% of all patients with response data and next-generation sequencing (NGS) data regardless of tumor type. *RB1* mutation was found in 15% of patients (Figure 3).
- No clear correlation was observed between genomic markers and best objective response (BOR).

Figure 3. Biomarker analysis. A) All available patients with NGS and response data (n=76); B) cohort 4.2 (n=11); C) cohort 7.4 (n=7)

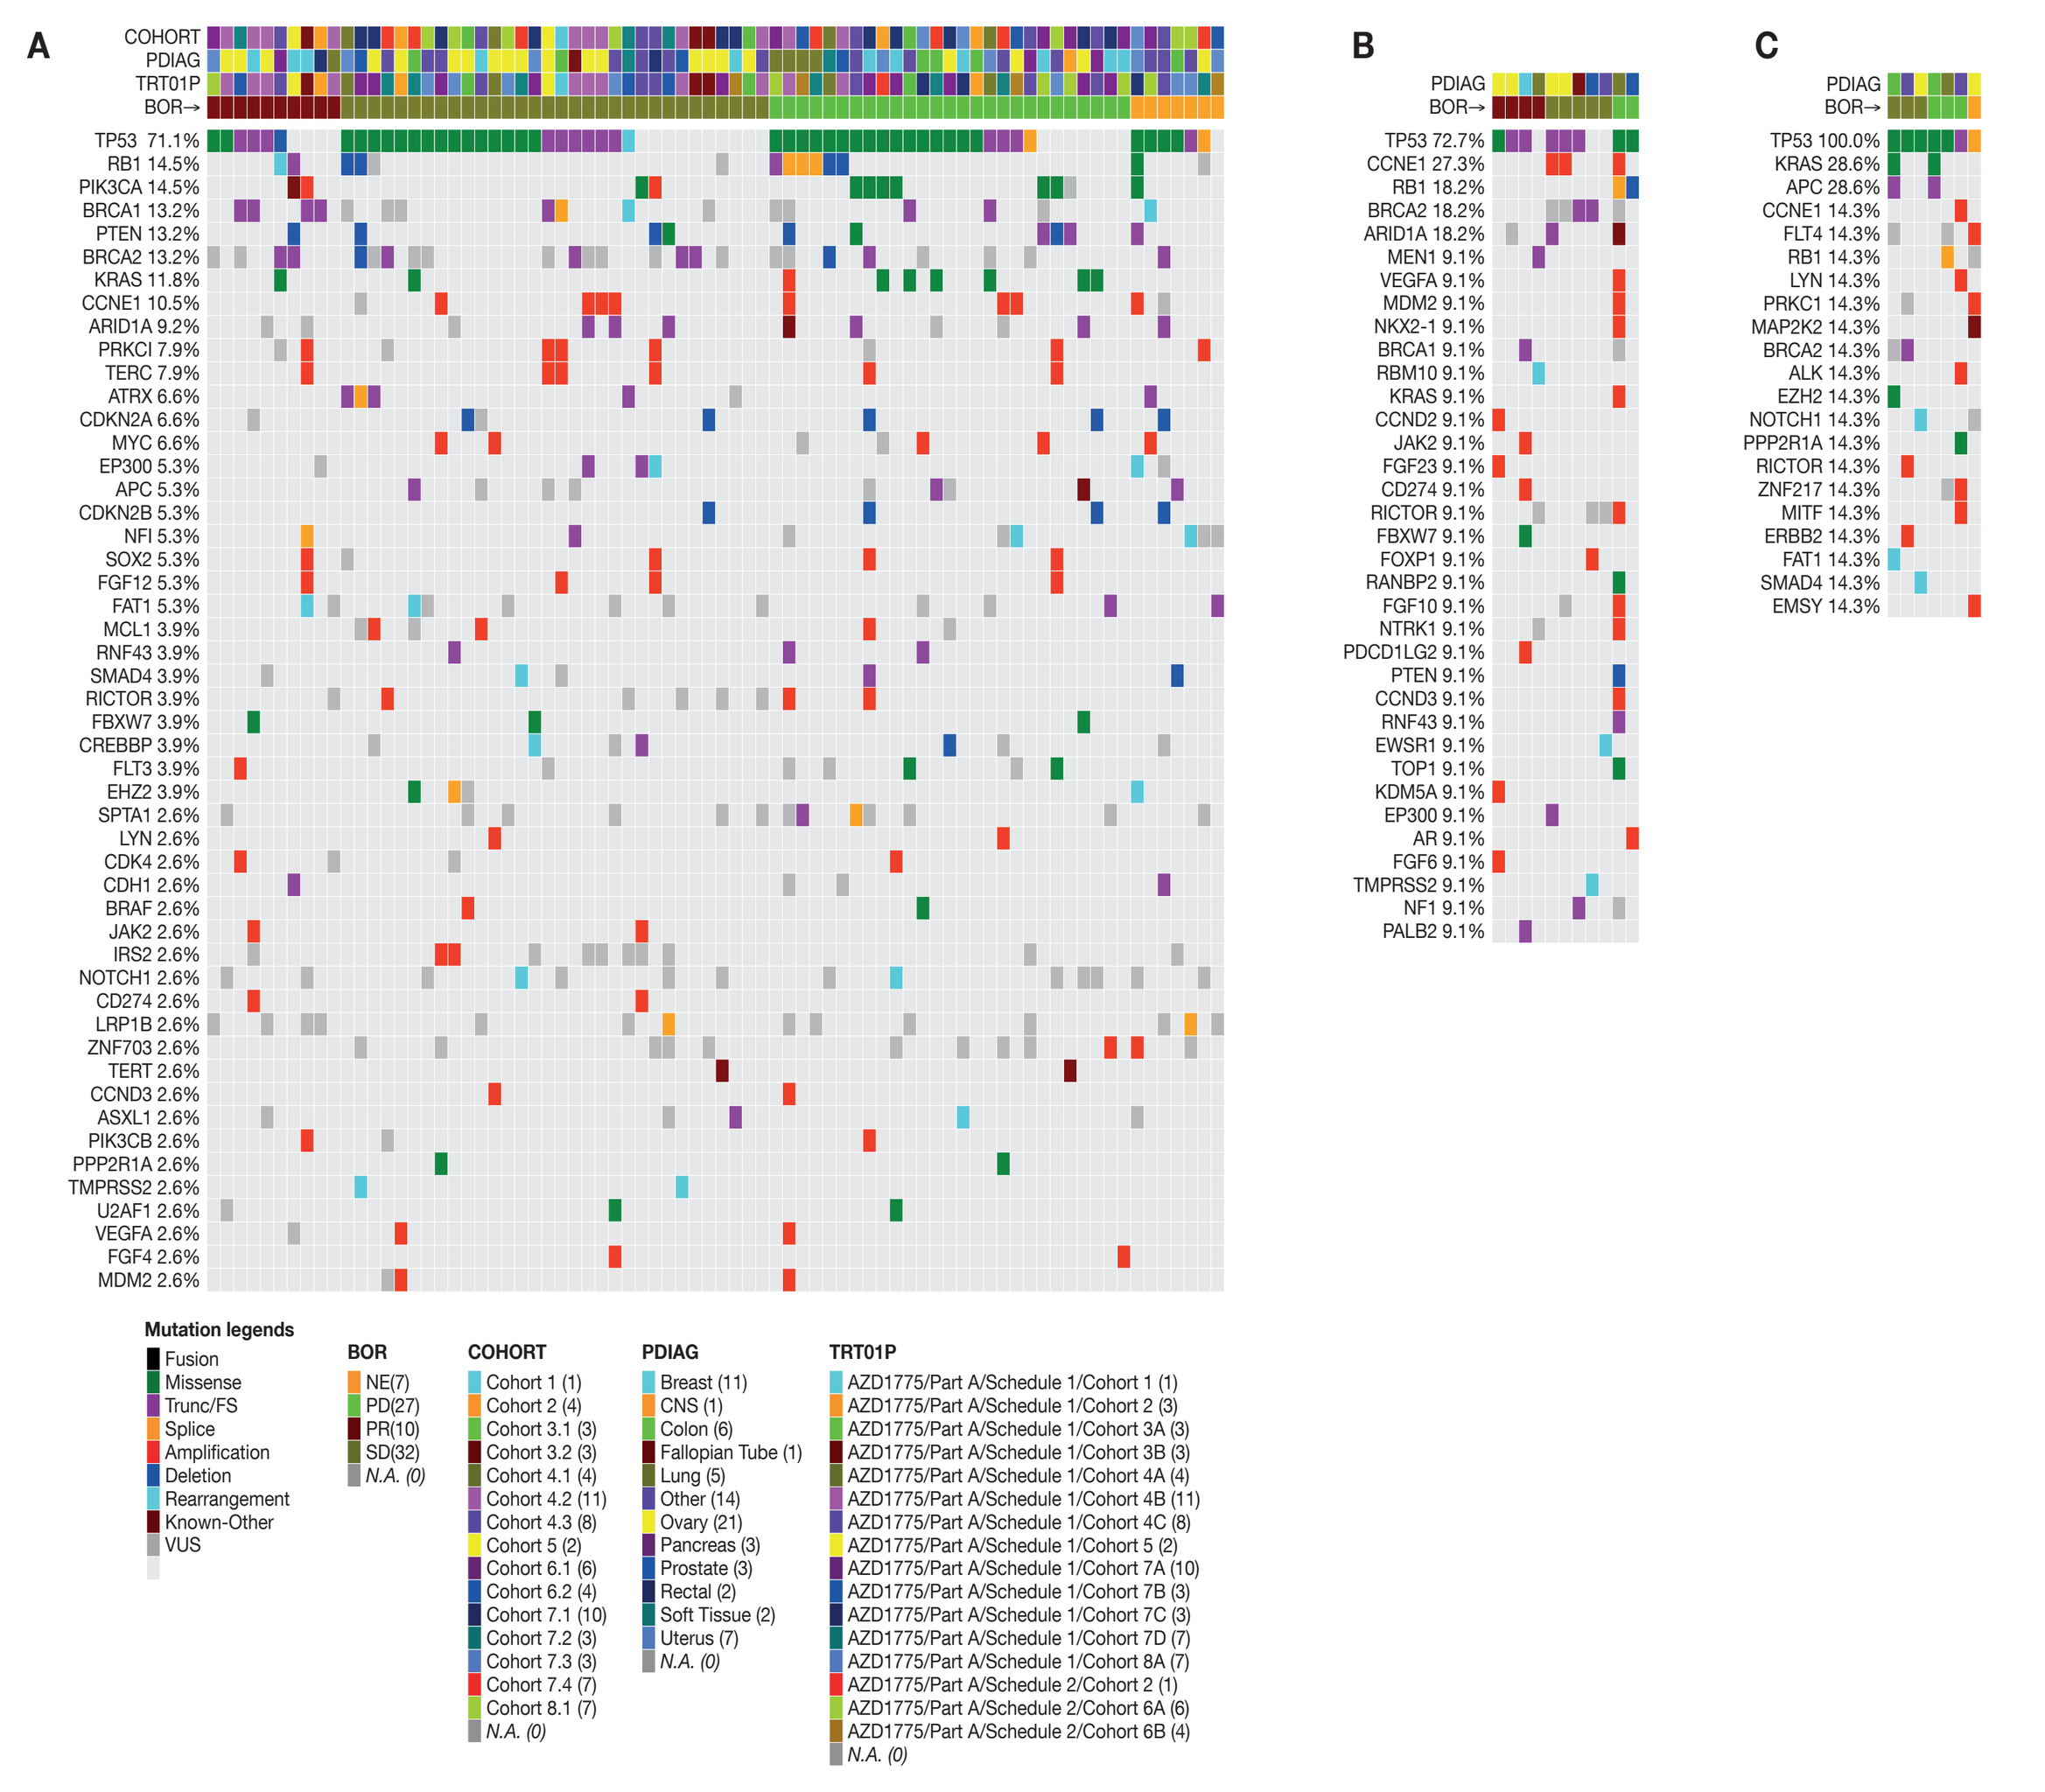
A
B
C
Mutation legends
Fusion
Missense
Trunc/FS
Splice
Amplification
Deletion
Rearrangement
Known-Other
VUS
BOR
COHORT
PDIAG
TRT01P

Conclusions

- The findings of this study are consistent with the known safety profile for adavosertib. Overall, the safety and tolerability profile was considered manageable for the patient population for which standard-of-care therapy does not exist or has proven ineffective or intolerable.
- The MTD/RP2D for the bid schedule was determined to be adavosertib 175 mg bid (3/4) for 2/3 weeks plus olaparib 200 mg bid. This was subsequently changed to adavosertib 150 mg bid plus olaparib 200 mg bid because of safety signals observed within a Phase II study.
- The RP2D for the qd schedule was adavosertib 200 mg qd (3/4) for 2/3 weeks plus olaparib 200 mg bid; this is being carried forward into the dose-expansion part of this study, in which the combination of adavosertib and olaparib will be tested in patients with small-cell lung cancer.
- PK analysis showed that adavosertib 175 mg bid (3/4) provided the greatest cover during the dosing interval and that exposure to olaparib is extended with combination treatment.
- Antitumor activity was demonstrated with adavosertib in combination with olaparib in patients with BRCA-mutant and wild-type cancers.
- No clear correlation between genomic profile and clinical response was observed, but analyses for biomarkers predicting the response seen in this study are ongoing.

References

1. Do K *et al. J Clin Oncol* 2015;33:3406–3415.
2. Lin X *et al. J Exp Clin Cancer Res* 2018;37:129.
3. Meng X *et al. Cancers (Basel)* 2018;10:E149.
4. Ledermann J *et al. N Engl J Med* 2012;366:1382–1392.
5. Kaufman B *et al. J Clin Oncol* 2015;33:244–250.
6. O'Connor MJ *et al. Eur J Cancer* 2016;69(Suppl 1):abst 436.

Acknowledgments

This study was sponsored by AstraZeneca. Medical writing assistance was provided by Jonathan Morton, PhD, from Comradis, funded by AstraZeneca. The authors would like to thank all participating patients and their families.

www.astrazeneca-medimmuneoncologycongresses.com/0ohjax
Copies of this poster obtained through QR (Quick Response) code are for pesonal use only and may not be reproduced without permission from the authors of this poster

Presented at AACR Annual Meeting, March 29 – April 3, 2019, Atlanta, GA, USA